A Novel Oral Pyruvate Kinase M2 (PKM2) Modulator for Atopic Dermatitis (SYX-5219) Demonstrates Favorable Safety, Pharmacokinetics, Robust Target Engagement and Evidence of Type 2 Immunomodulation: Results from a First-in-Human Phase 1a Study

Alan Irvine[1], Raj Chovatiya[2], Lisa Pierre[3], Niamh Richmond[3], Tom Ashton[3], Kevin Duffy[3], Natalie Jones[3], Gordon Dingwall[3], Iain Kilty[3], Ravi Rao[3]

1. St James's Hospital, Dublin, Leinster, Ireland.
2. Center for Medical Dermatology + Immunology Research, Chicago, IL, United States.
3. Sitryx Therapeutics Ltd, 101 Bellhouse, Oxford Science Park, Oxford, United Kingdom.

Introduction and background

SYX-5219 promotes PKM2 tetramerization to reduce inflammation

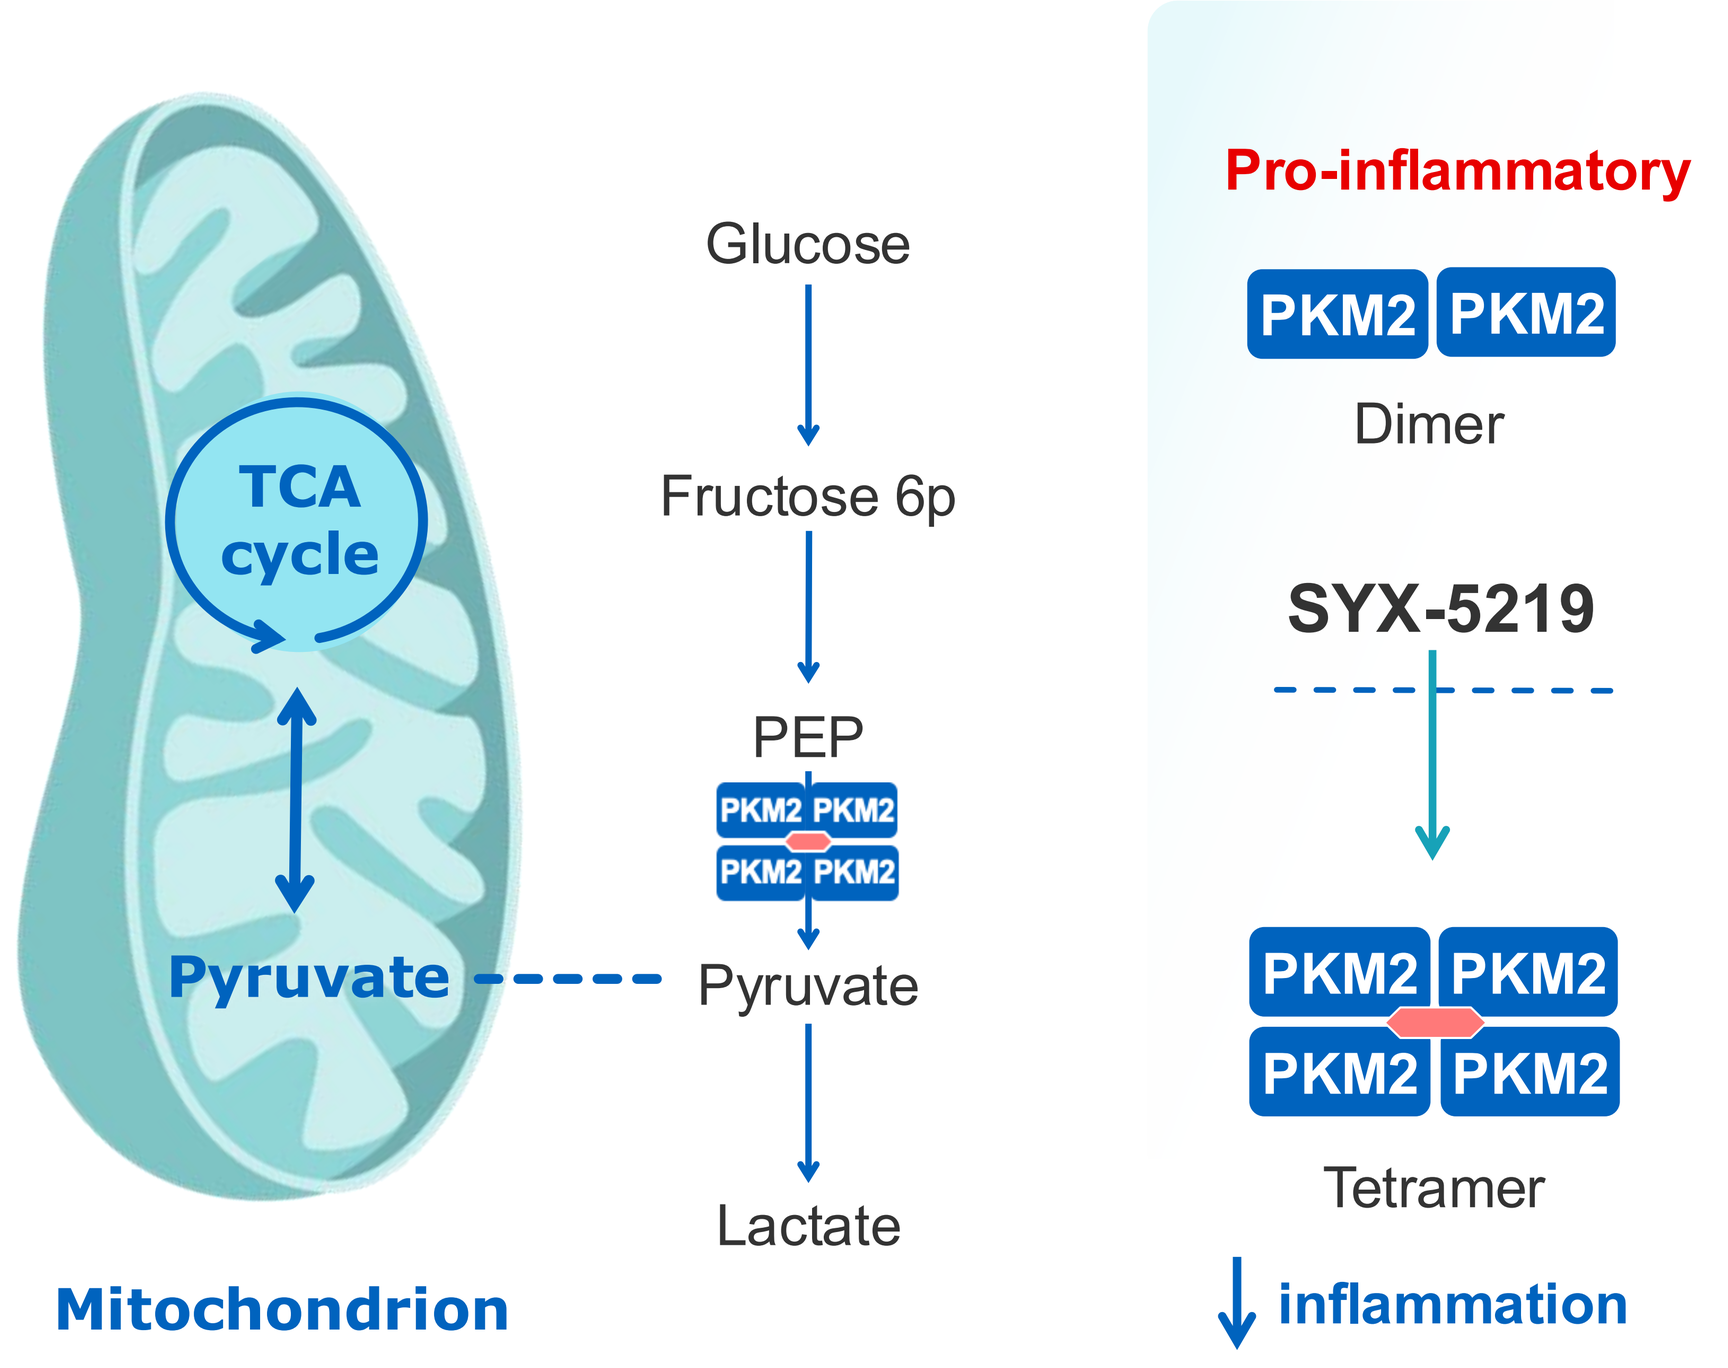

TCA cycle
Pyruvate
Mitochondrion
Glucose
Fructose 6p
PEP
Pyruvate
Lactate
Pro-inflammatory
PKM2 PKM2
Dimer
SYX-5219
PKM2 PKM2
PKM2 PKM2
Tetramer
↓ inflammation

PKM2 is upregulated in AD skin
Representative PKM2 immunohistochemistry (IHC) staining of healthy and severe AD lesional skin

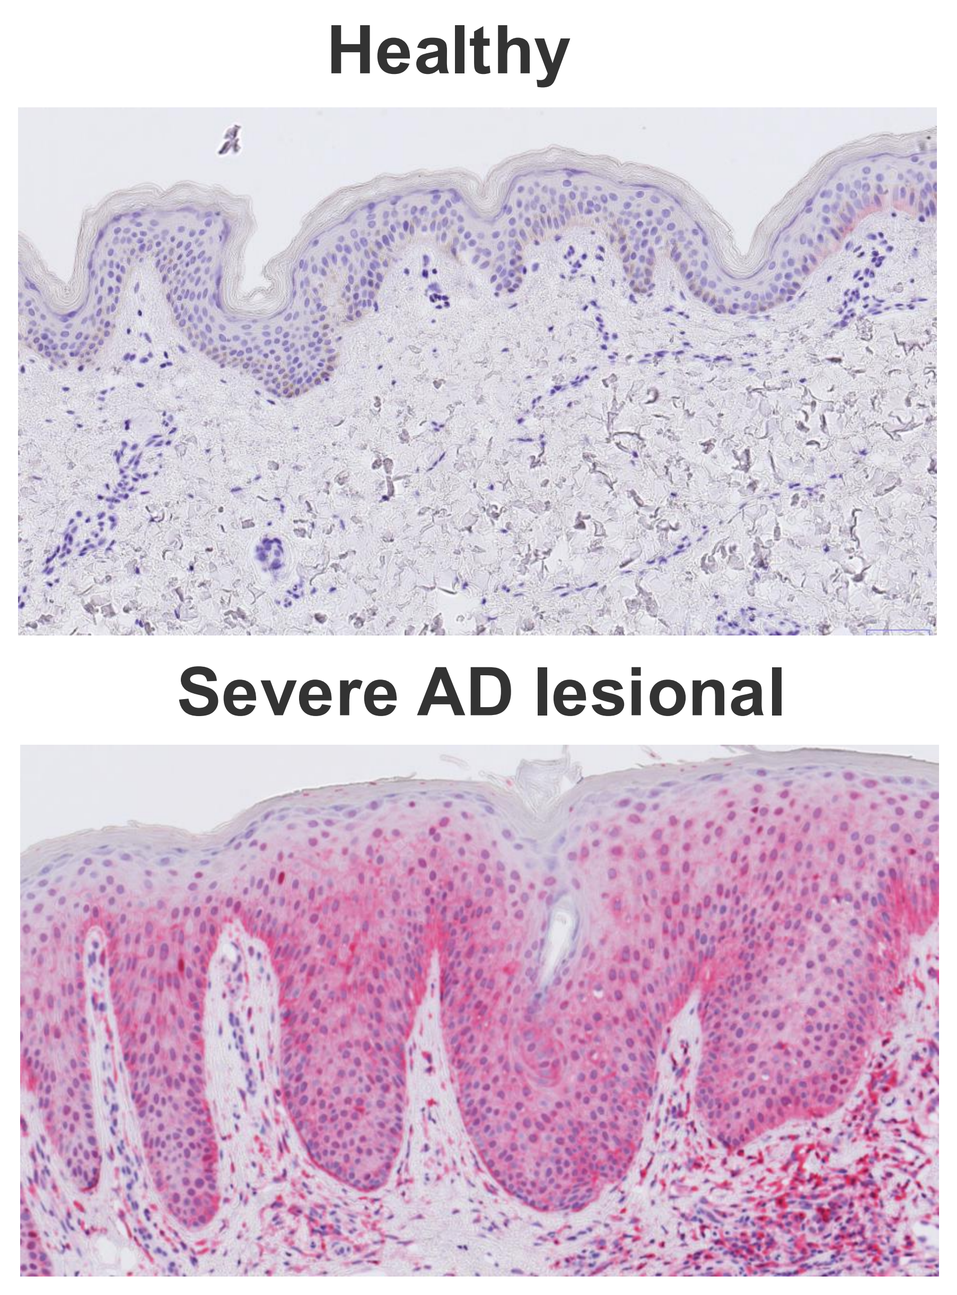

Healthy
Severe AD lesional

Modulation of PKM2 impacts multiple drivers of atopic dermatitis (AD) pathology

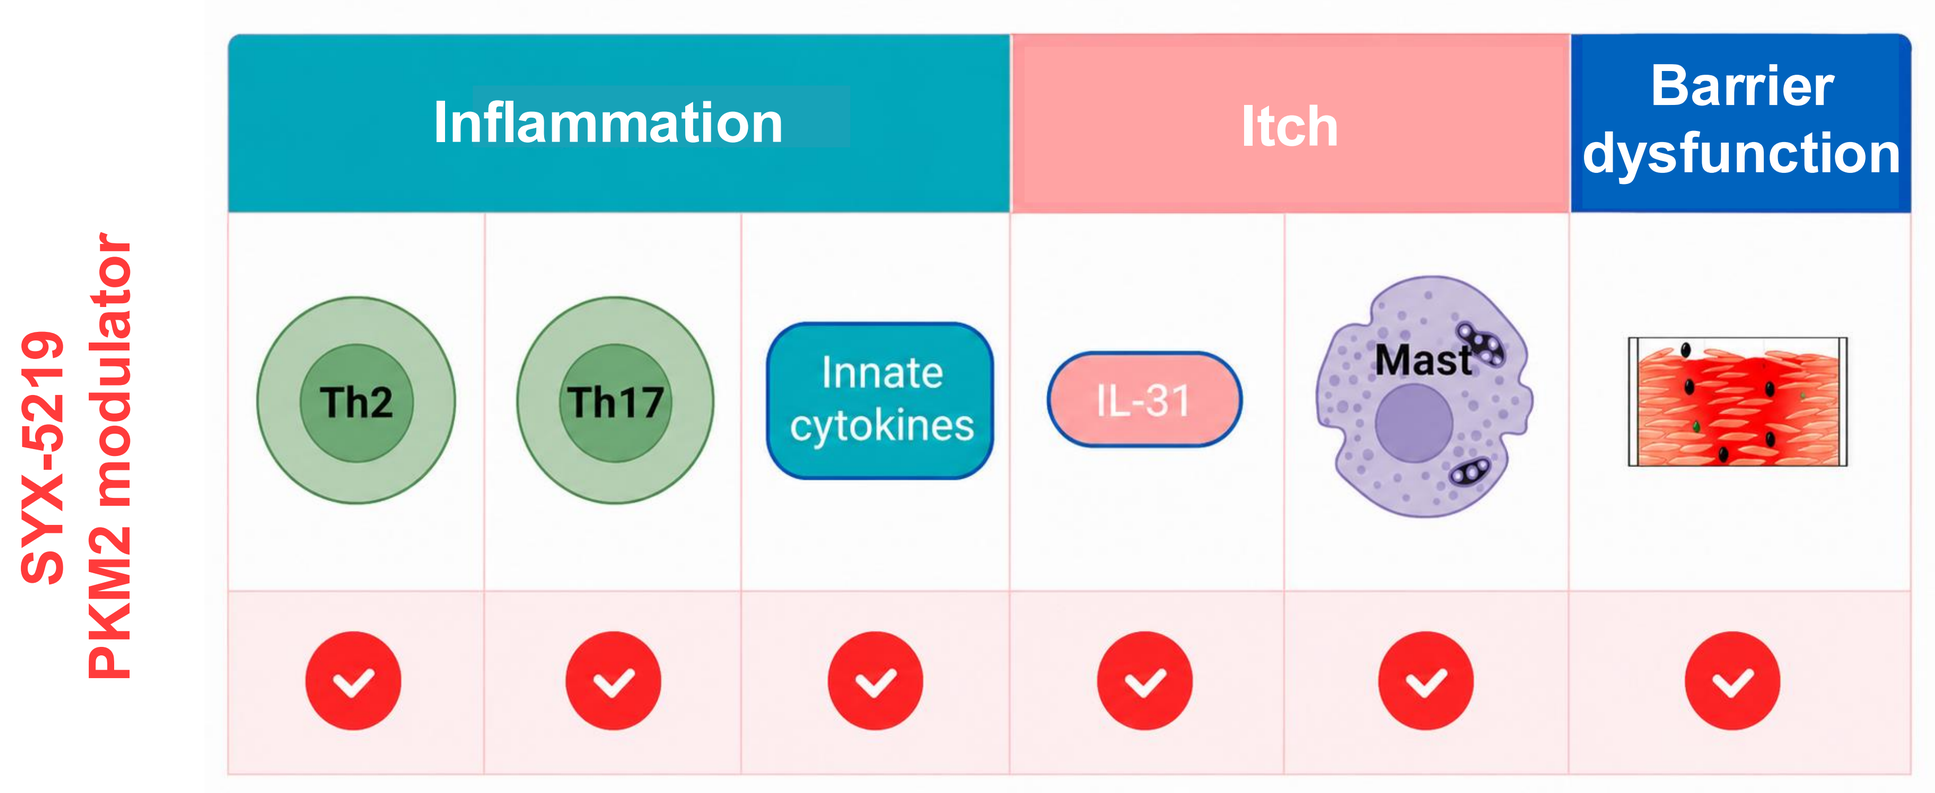

SYX-5219 PKM2 modulator
Inflammation
Itch
Barrier dysfunction
Th2
Th17
Innate cytokines
IL-31
Mast

Methods

- First-in-human, placebo-controlled study of oral SYX-5219 in healthy volunteers (aged 18-65y)
- 96 participants exposed overall; 72 received SYX-5219 (24 received placebo)
- Assessments included safety/tolerability, PK, PBMC pyruvate kinase activity, and serum TARC/CCL17

Study design

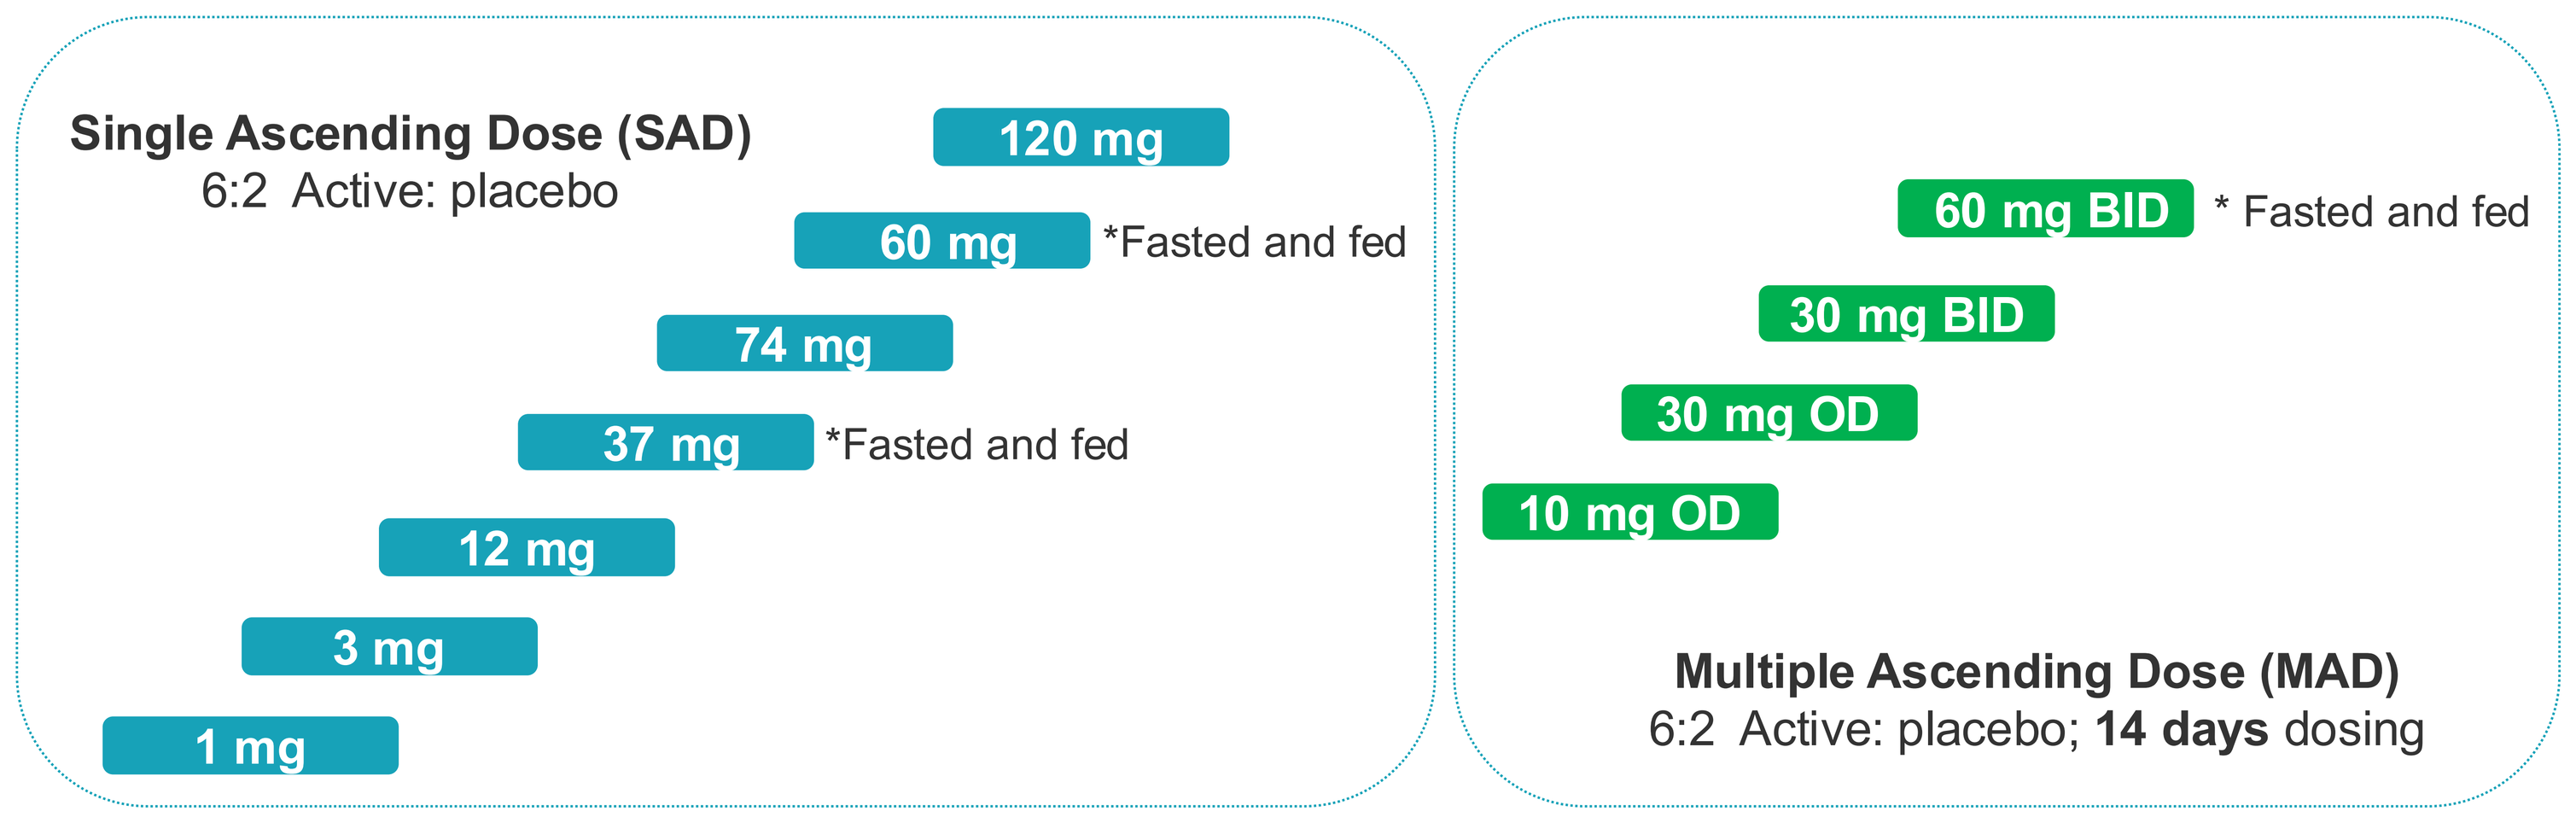

Single Ascending Dose (SAD)
6:2 Active: placebo
120 mg
60 mg *Fasted and fed
74 mg
37 mg *Fasted and fed
12 mg
3 mg
1 mg
60 mg BID * Fasted and fed
30 mg BID
30 mg OD
10 mg OD
Multiple Ascending Dose (MAD)
6:2 Active: placebo; 14 days dosing

Results

Safety and tolerability

SYX-5219 was **safe and well tolerated** across all SAD and 14-day MAD cohorts in healthy adult participants. No serious or severe adverse events were reported, and no treatment-related withdrawals or laboratory abnormalities were observed.

n (%)	Placebo Overall (N=10)	SYX-5219 Overall (N=30)	10 mg QD		30 mg QD		30 mg BID		60 mg BID	
			Placebo (N=2)	SYX-5219 (N=6)	Placebo (N=2)	SYX-5219 (N=6)	Placebo (N=2)	SYX-5219 (N=6)	Placebo (N=4)	SYX-5219 (N=12)
Any TEAE	3 (30.0)	11 (36.7)	0	0	0	2 (33.3)	1 (50.0)	2 (33.3)	2 (50.0)	7 (58.3)
Serious TEAE	0	0	0	0	0	0	0	0	0	0
Mild	3 (30.0)	5 (16.7)	0	0	0	0	1 (50.0)	2 (33.3)	2 (50.0)	3 (25.0)
Moderate	0	6 (20.0)	0	0	0	2 (33.3)	0	0	0	4 (33.3)
Severe	0	0	0	0	0	0	0	0	0	0

Pharmacokinetics

- SYX-5219 showed sustained Day 14 plasma exposure across MAD cohorts.
- Single-dose exposure was dose-proportional up to 60 mg.
- Apparent half-life was ~60-70 h, with steady state estimated by Day 6-8.
- No food effect was observed at steady state.
- Steady state exposure consistent with preclinical human dose prediction.

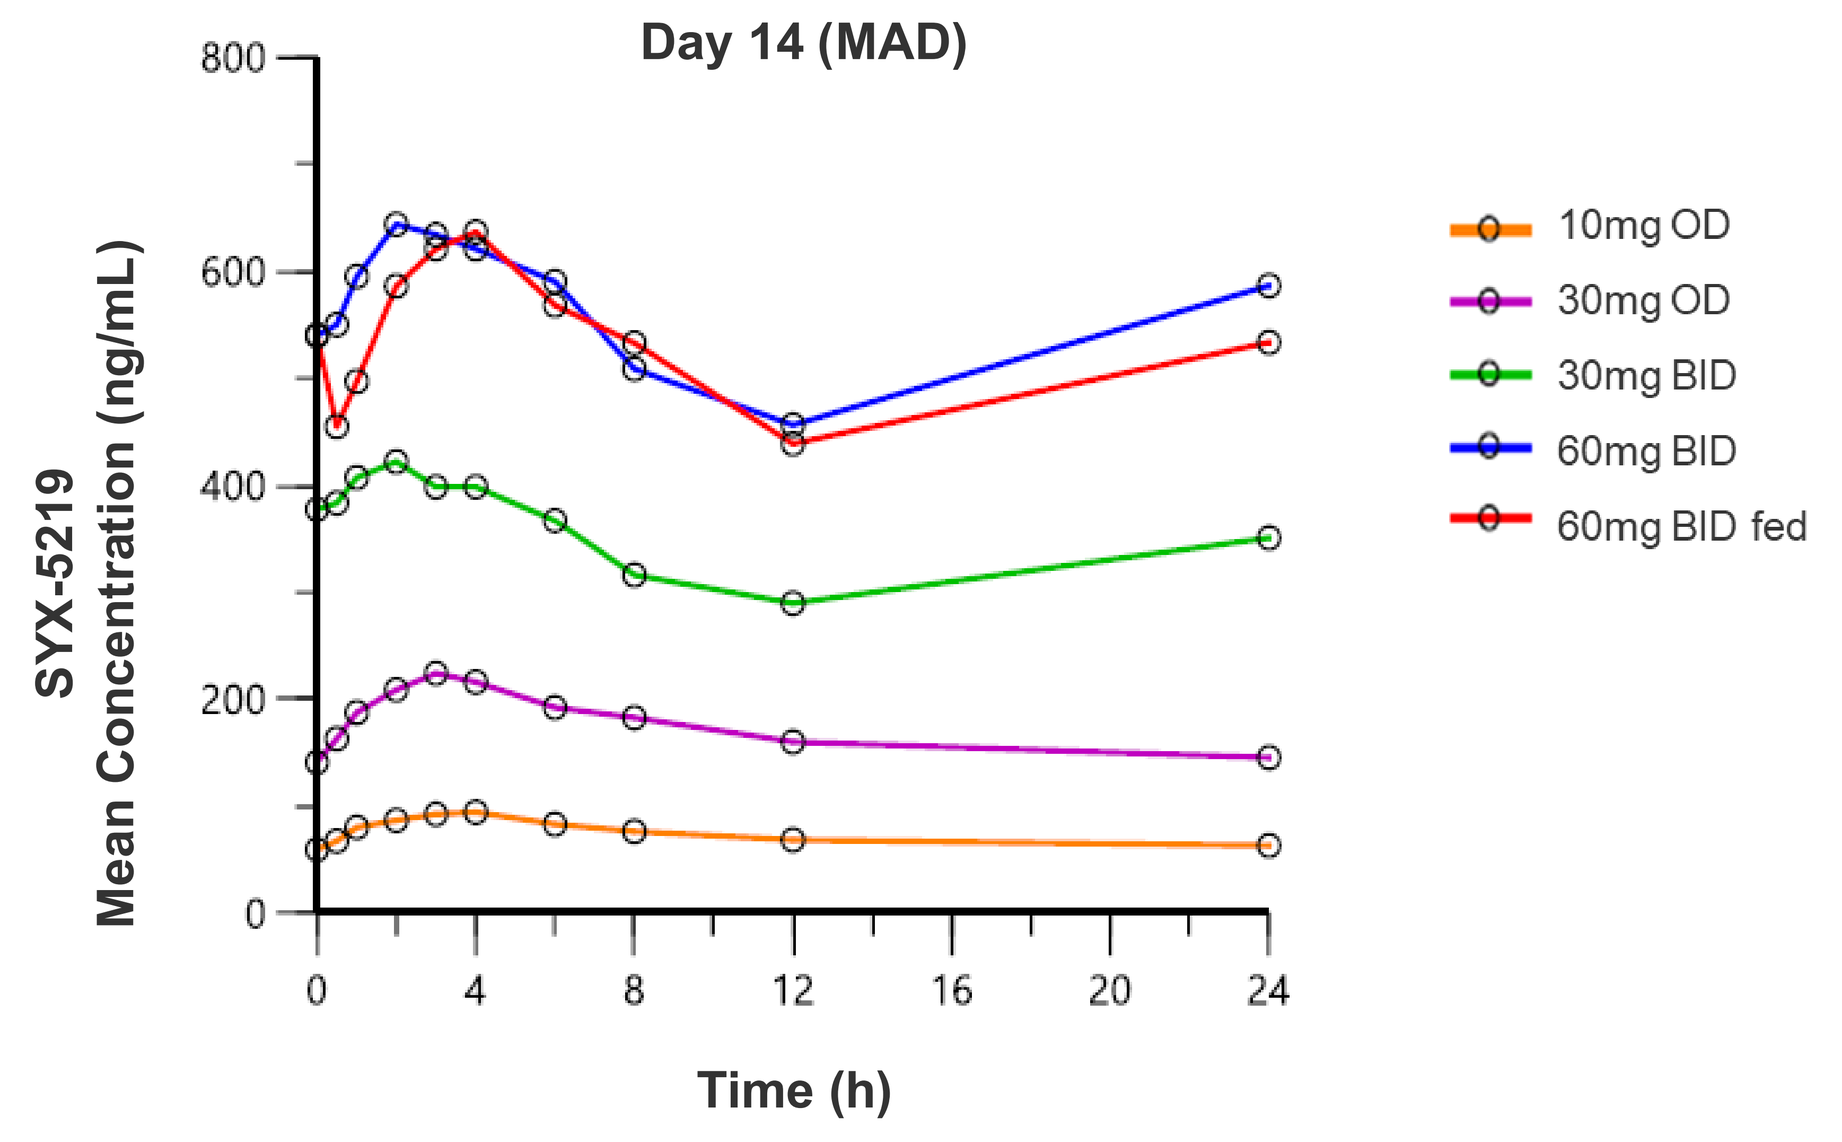

Day 14 (MAD)
SYX-5219 Mean Concentration (ng/mL)
Time (h)
10mg OD
30mg OD
30mg BID
60mg BID
60mg BID fed

Target engagement and Pharmacodynamics

SYX-5219 achieved exposure-dependent pyruvate kinase target engagement

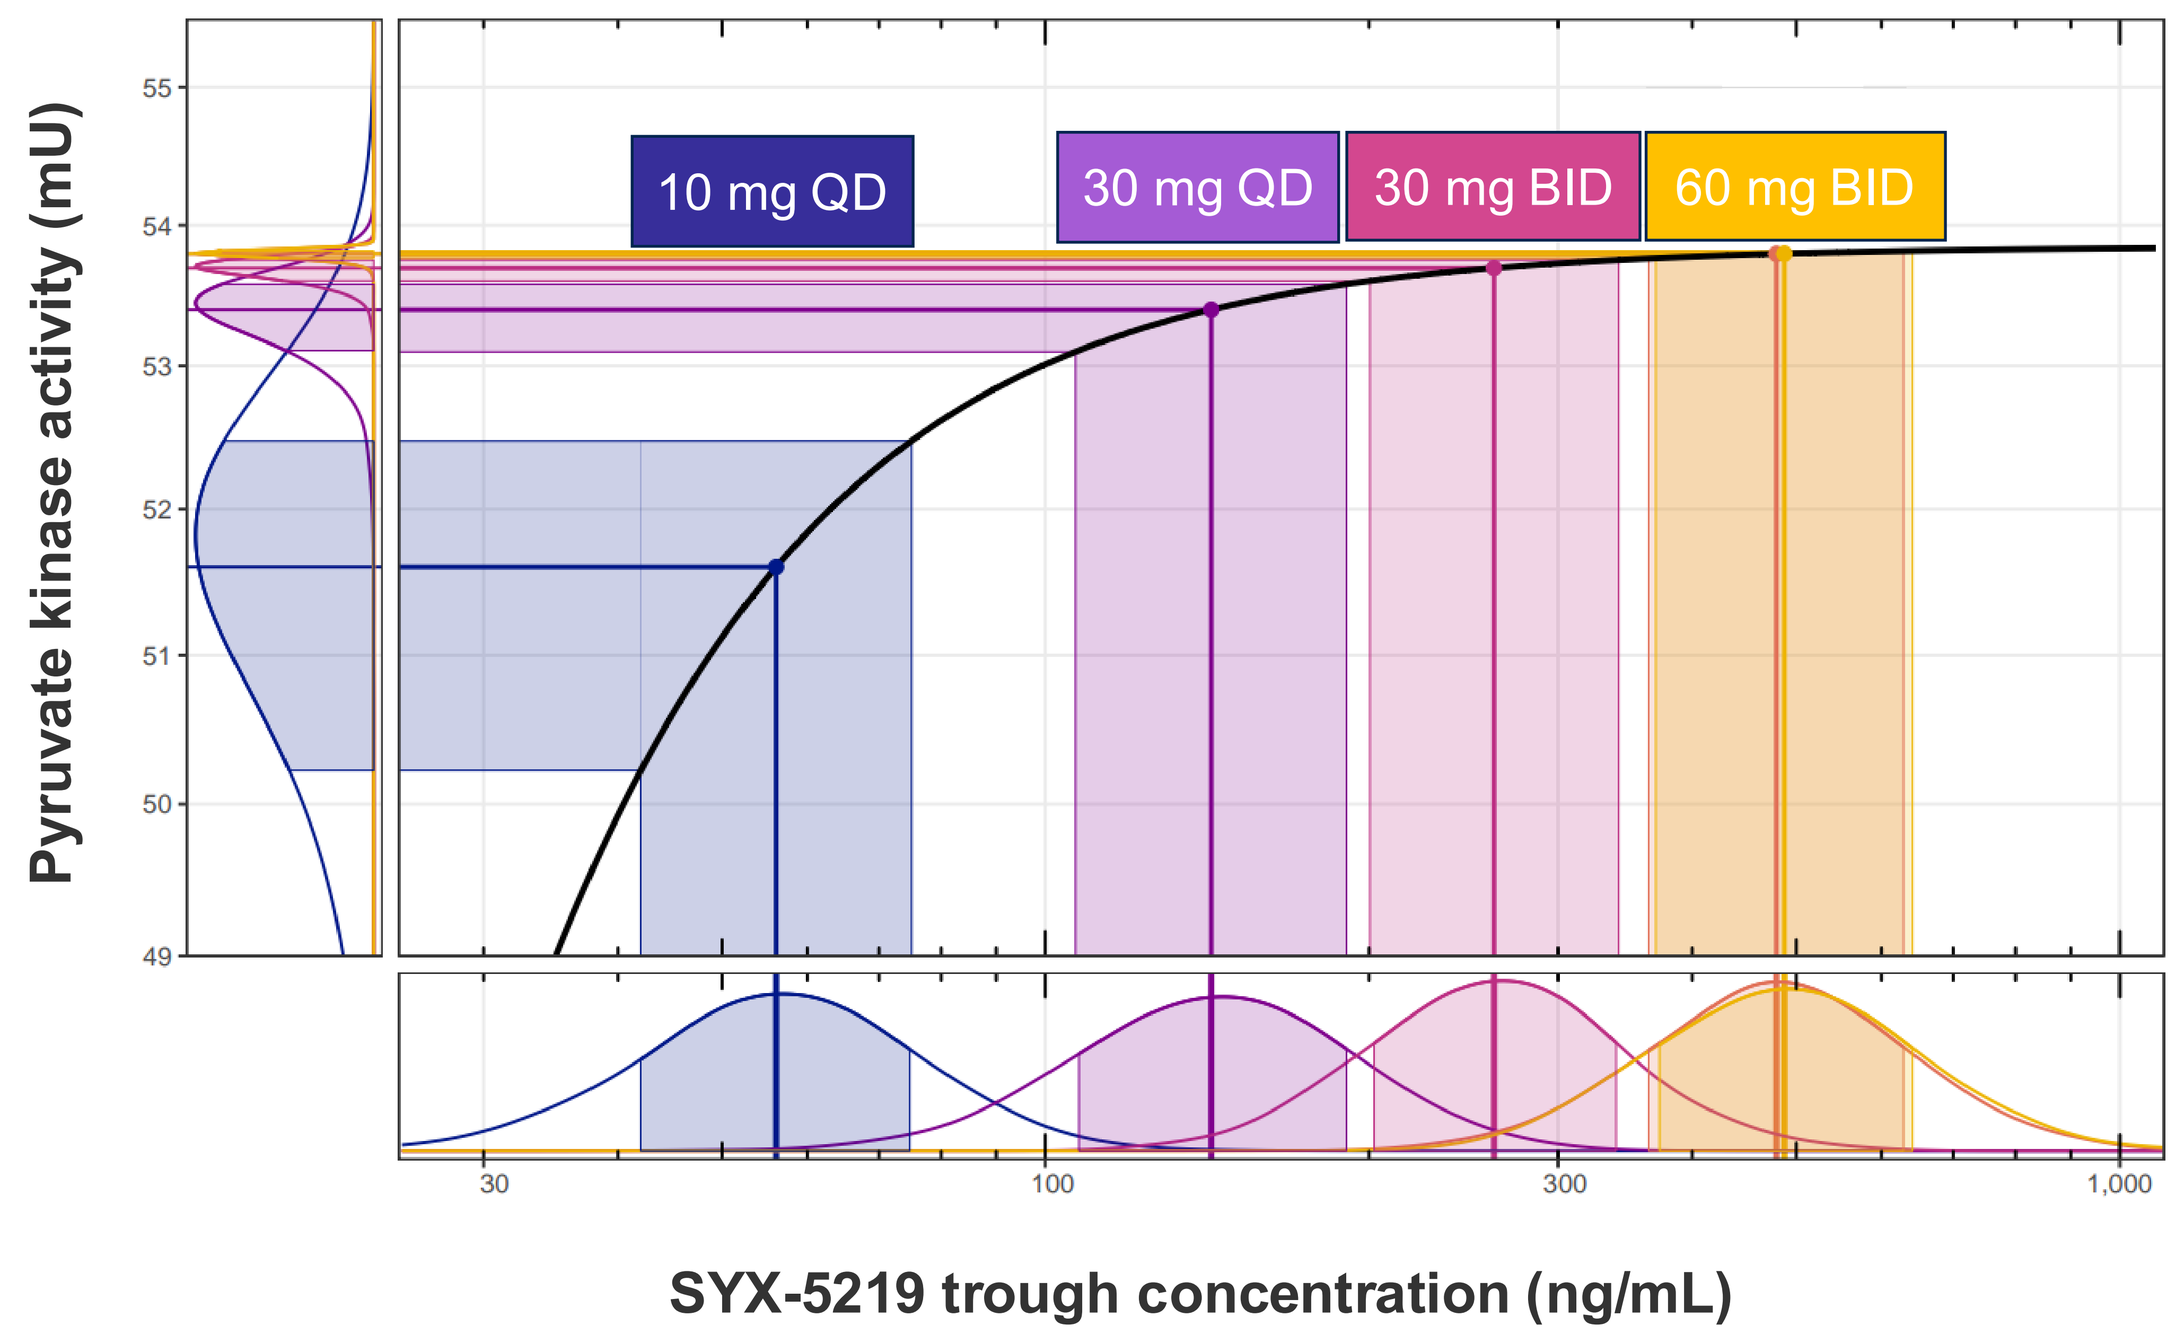

Pyruvate kinase activity (mU)
10 mg QD
30 mg QD
30 mg BID
60 mg BID
SYX-5219 trough concentration (ng/mL)

- Pyruvate kinase activity increased with SYX-5219 exposure and plateaued at higher plasma concentrations.
- Model-derived PK/PD analysis indicated ≥EC95 target engagement at doses ≥30 mg QD.
- BID dosing maintained exposures in the near-maximal target engagement range.
- These findings support robust pharmacologic engagement of PKM2 in humans.

SYX-5219 reduced AD inflammatory biomarker TARC/CCL17 in healthy adults

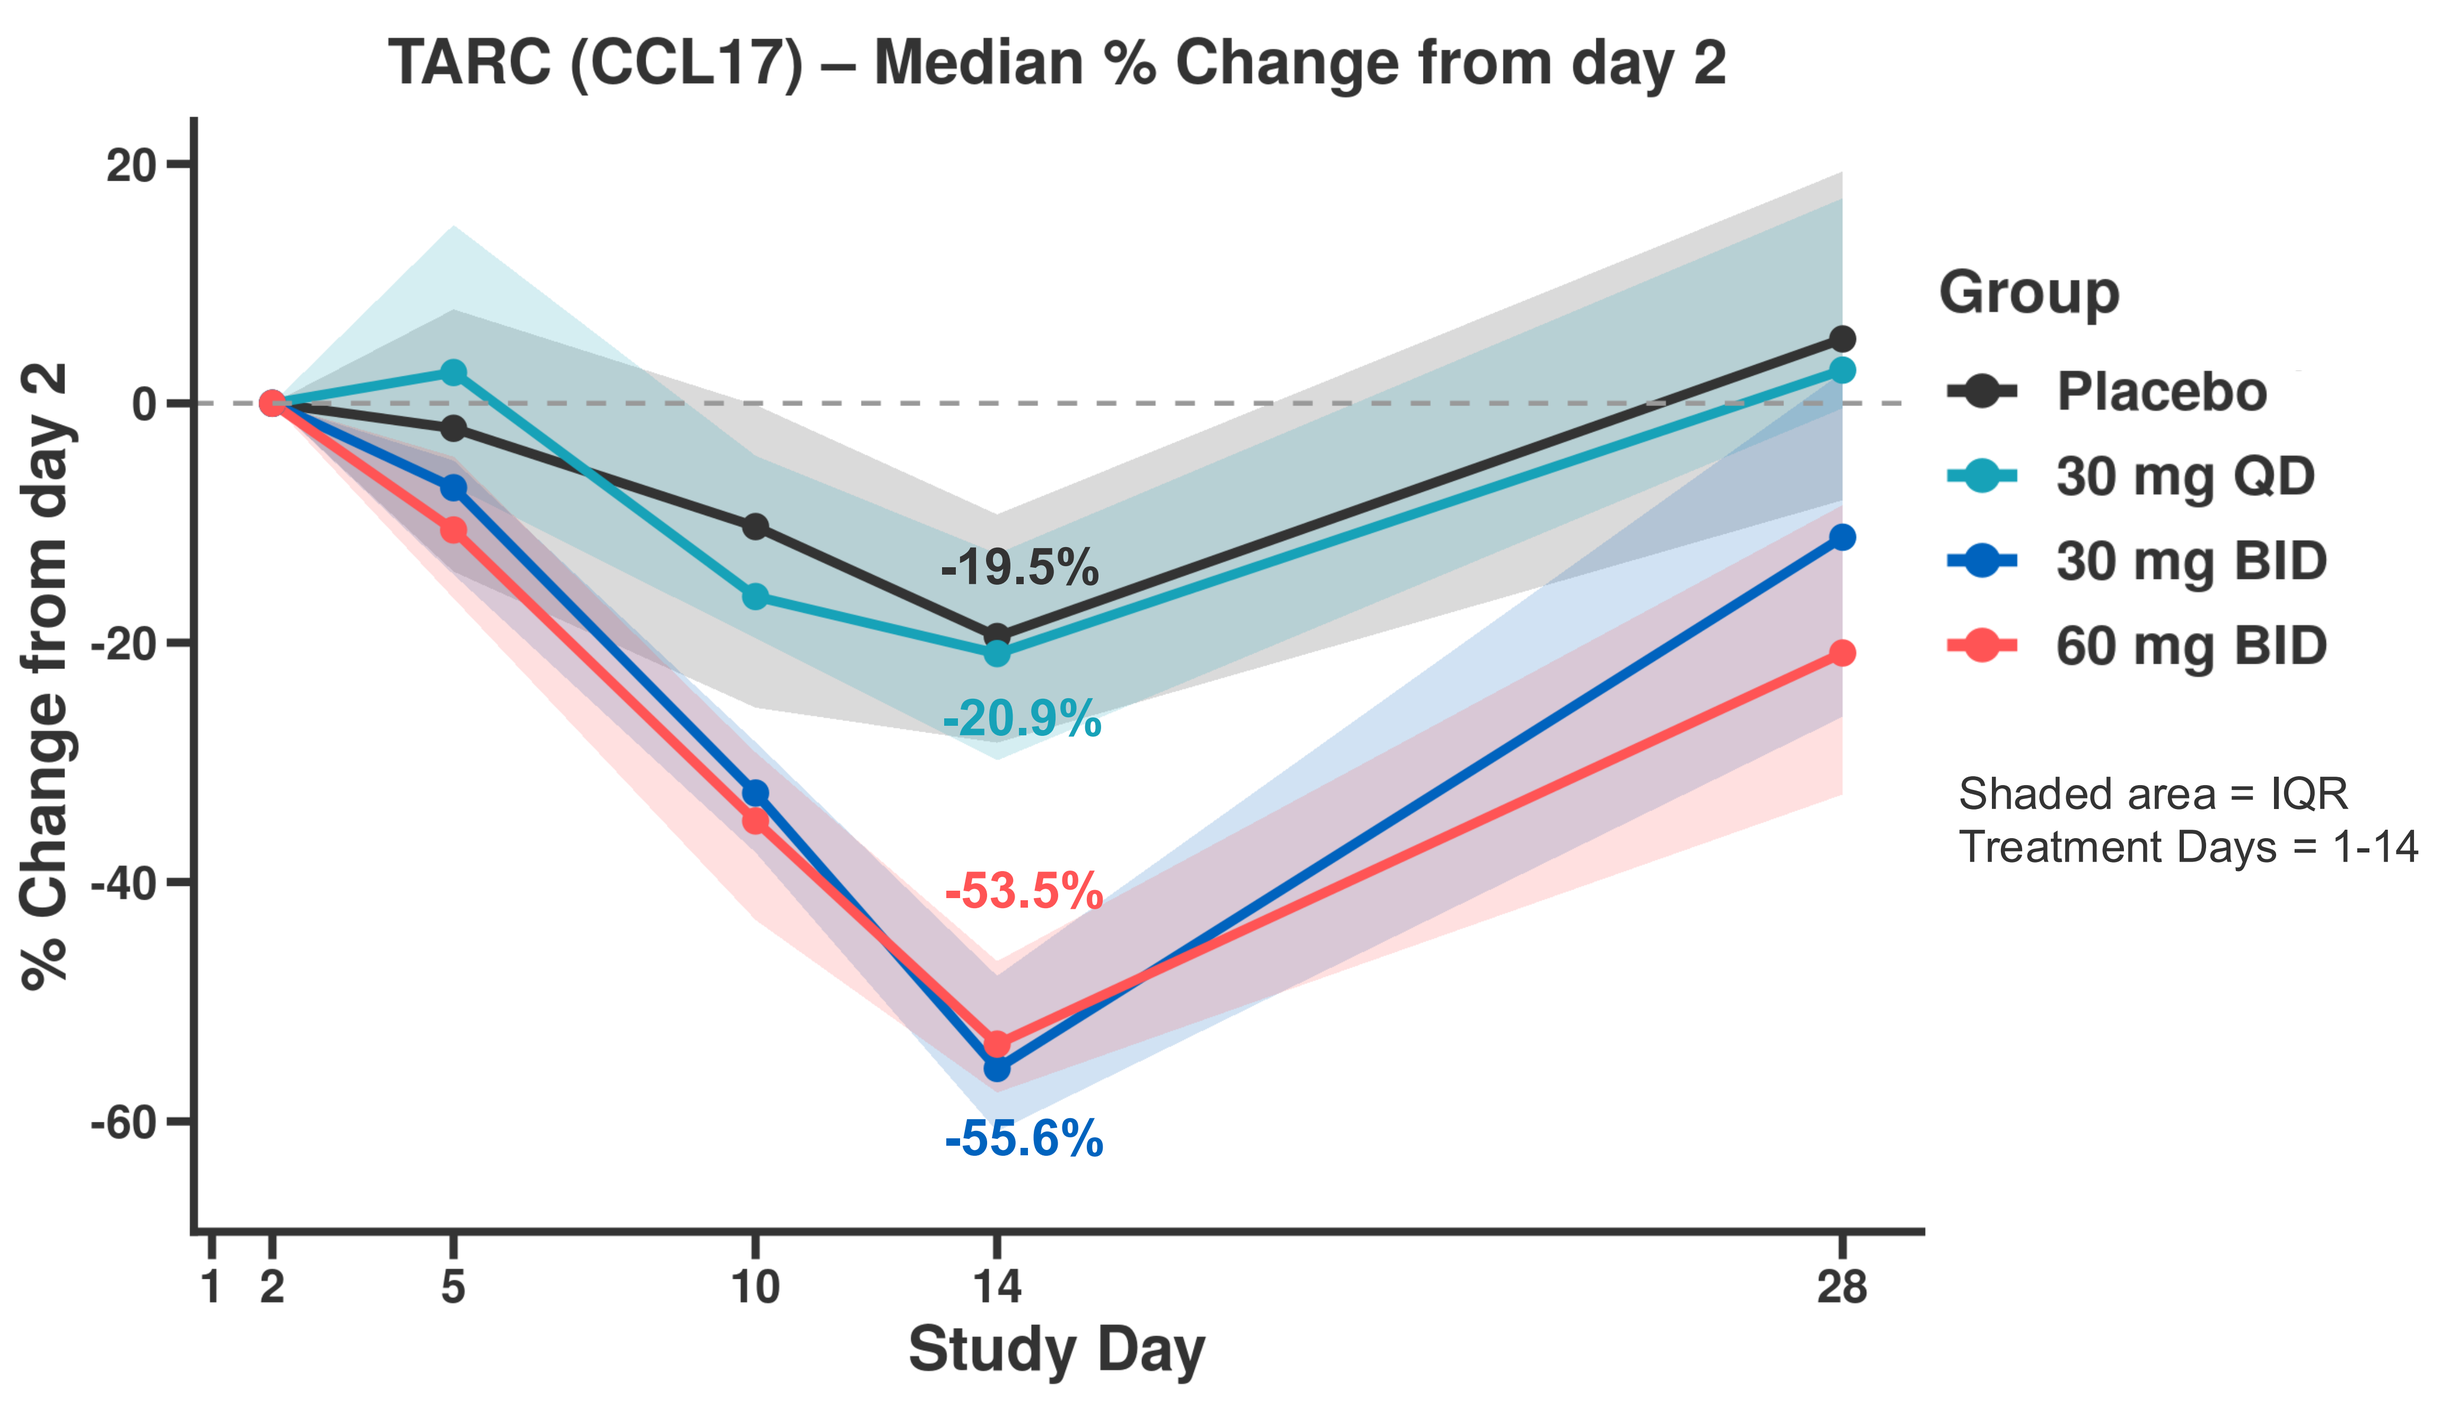

TARC (CCL17) – Median % Change from day 2
% Change from day 2
Study Day
-19.5%
-20.9%
-53.5%
-55.6%
Group
Placebo
30 mg QD
30 mg BID
60 mg BID
Shaded area = IQR
Treatment Days = 1-14

- SYX-5219 reduced serum TARC/CCL17 during 14-day dosing.
- Greatest median reductions were observed at Day 14 in BID cohorts: −55.6% with 30 mg BID and −53.5% with 60 mg BID.
- TARC/CCL17 levels trended back towards normalising at follow-up (Day 28) after treatment cessation.
- These data support systemic modulation of a type 2 inflammatory biomarker

Conclusions

SYX-5219, a first-in-class oral PKM2 modulator, demonstrated a favorable Phase 1a profile in healthy adults supportive of progression into a Phase 1b study in AD.
Across SAD and 14-day MAD cohorts, SYX-5219 was safe and well tolerated, achieved sustained systemic exposures, and showed robust exposure-dependent target engagement. Treatment was associated with reductions in serum TARC/CCL17, supporting systemic modulation of type 2 inflammatory biomarkers. Together, these data support further clinical evaluation of SYX-5219 as a novel immunometabolic approach for AD.

Future direction

An ongoing Phase 1b study is evaluating SYX-5219 in adults with moderate-to-severe AD.
The AD cohort will enroll up to 45 participants and assess safety, tolerability, PK, target engagement, clinical efficacy and disease-associated biomarkers following multiple dosing of SYX-5219 or placebo.

Acknowledgements

The authors thank the study participants and clinical site staff. In particular, Dr Annelize Koch and the Simbec-Orion team for their contributions to study conduct, as well as the Sitryx clinical, translational and biomarker teams.

Company Confidential © 2026 Sitryx