PKM2 is a promising novel target in atopic dermatitis

Tom Ashton[1], Sabine Broc[1], Gareth Davies[1], Martin Domville[1], Eva Palsson-McDermott[1,2], Oscar Barba[1], Mark Whitmarsh[1], Lisa Pierre[1], Ravi Rao[1], Luke O'Neill[1,2], Iain Kilty[1]
[1]Sitryx Therapeutics Ltd, 101 Bellhouse, Oxford Science Park, Oxford, UK OX4 4GA
[2]School of Biochemistry and Immunology, Trinity Biomedical Science Institute, Trinity College Dublin, Ireland

Introduction

Atopic dermatitis (AD) is a complex, chronic inflammatory, non-contagious skin disease with high unmet need and no curative treatment options. Pyruvate kinase M2 (PKM2) is a key enzyme in the central metabolic pathway of glycolysis and is specifically upregulated in inflammatory tissues. Here we describe PKM2 as a novel target in AD and demonstrate efficacy of novel PKM2 modulators in the DNFB and oxazolone induced contact dermatitis mouse models of inflammatory skin disease. Efficacy is also demonstrated in IgE-induced passive cutaneous anaphylaxis and cortistatin-14 induced mast cell mouse models. A Sitryx PKM2 modulator, SYX-5219, has recently entered into a clinical trial for AD.

Methods

PKM2 expression was studied in human healthy skin and AD skin using immunohistochemistry (PKM2 Ab Cell Signalling, 4053S) and RNAseq. The *in vitro* pharmacology of PKM2 modulators, SYX-PKM-A and SYX-PKM-B, was studied using biochemical assays (Bio-Techne, 7244-PK-020) and pyruvate kinase activity (Abcam assay, ab83432) in human CD4+ T cells. The *in vivo* pharmacology of PKM2 modulators was determined in murine models. 2,4-dinitrofluorobenzene (DNFB) or oxazolone were used to induce contact dermatitis. IgE or cortistatin-14 were used to induce mast cell degranulation. Efficacy was measured using ear thickness, excoriation score, RNAseq from ear tissue, cytokine analysis from ear tissue (IL-1β, IL-4, IL-6, IL-13, IFNγ, IL-17a, IL-31 Luminex) or skin Evan's blue quantification. ANOVA with Dunnett's post-test * $p < 0.05$, ** $p < 0.01$, *** $p < 0.001$, **** $p < 0.0001$

Results

1. Resolution of inflammation in atopic dermatitis with a PKM2 modulator

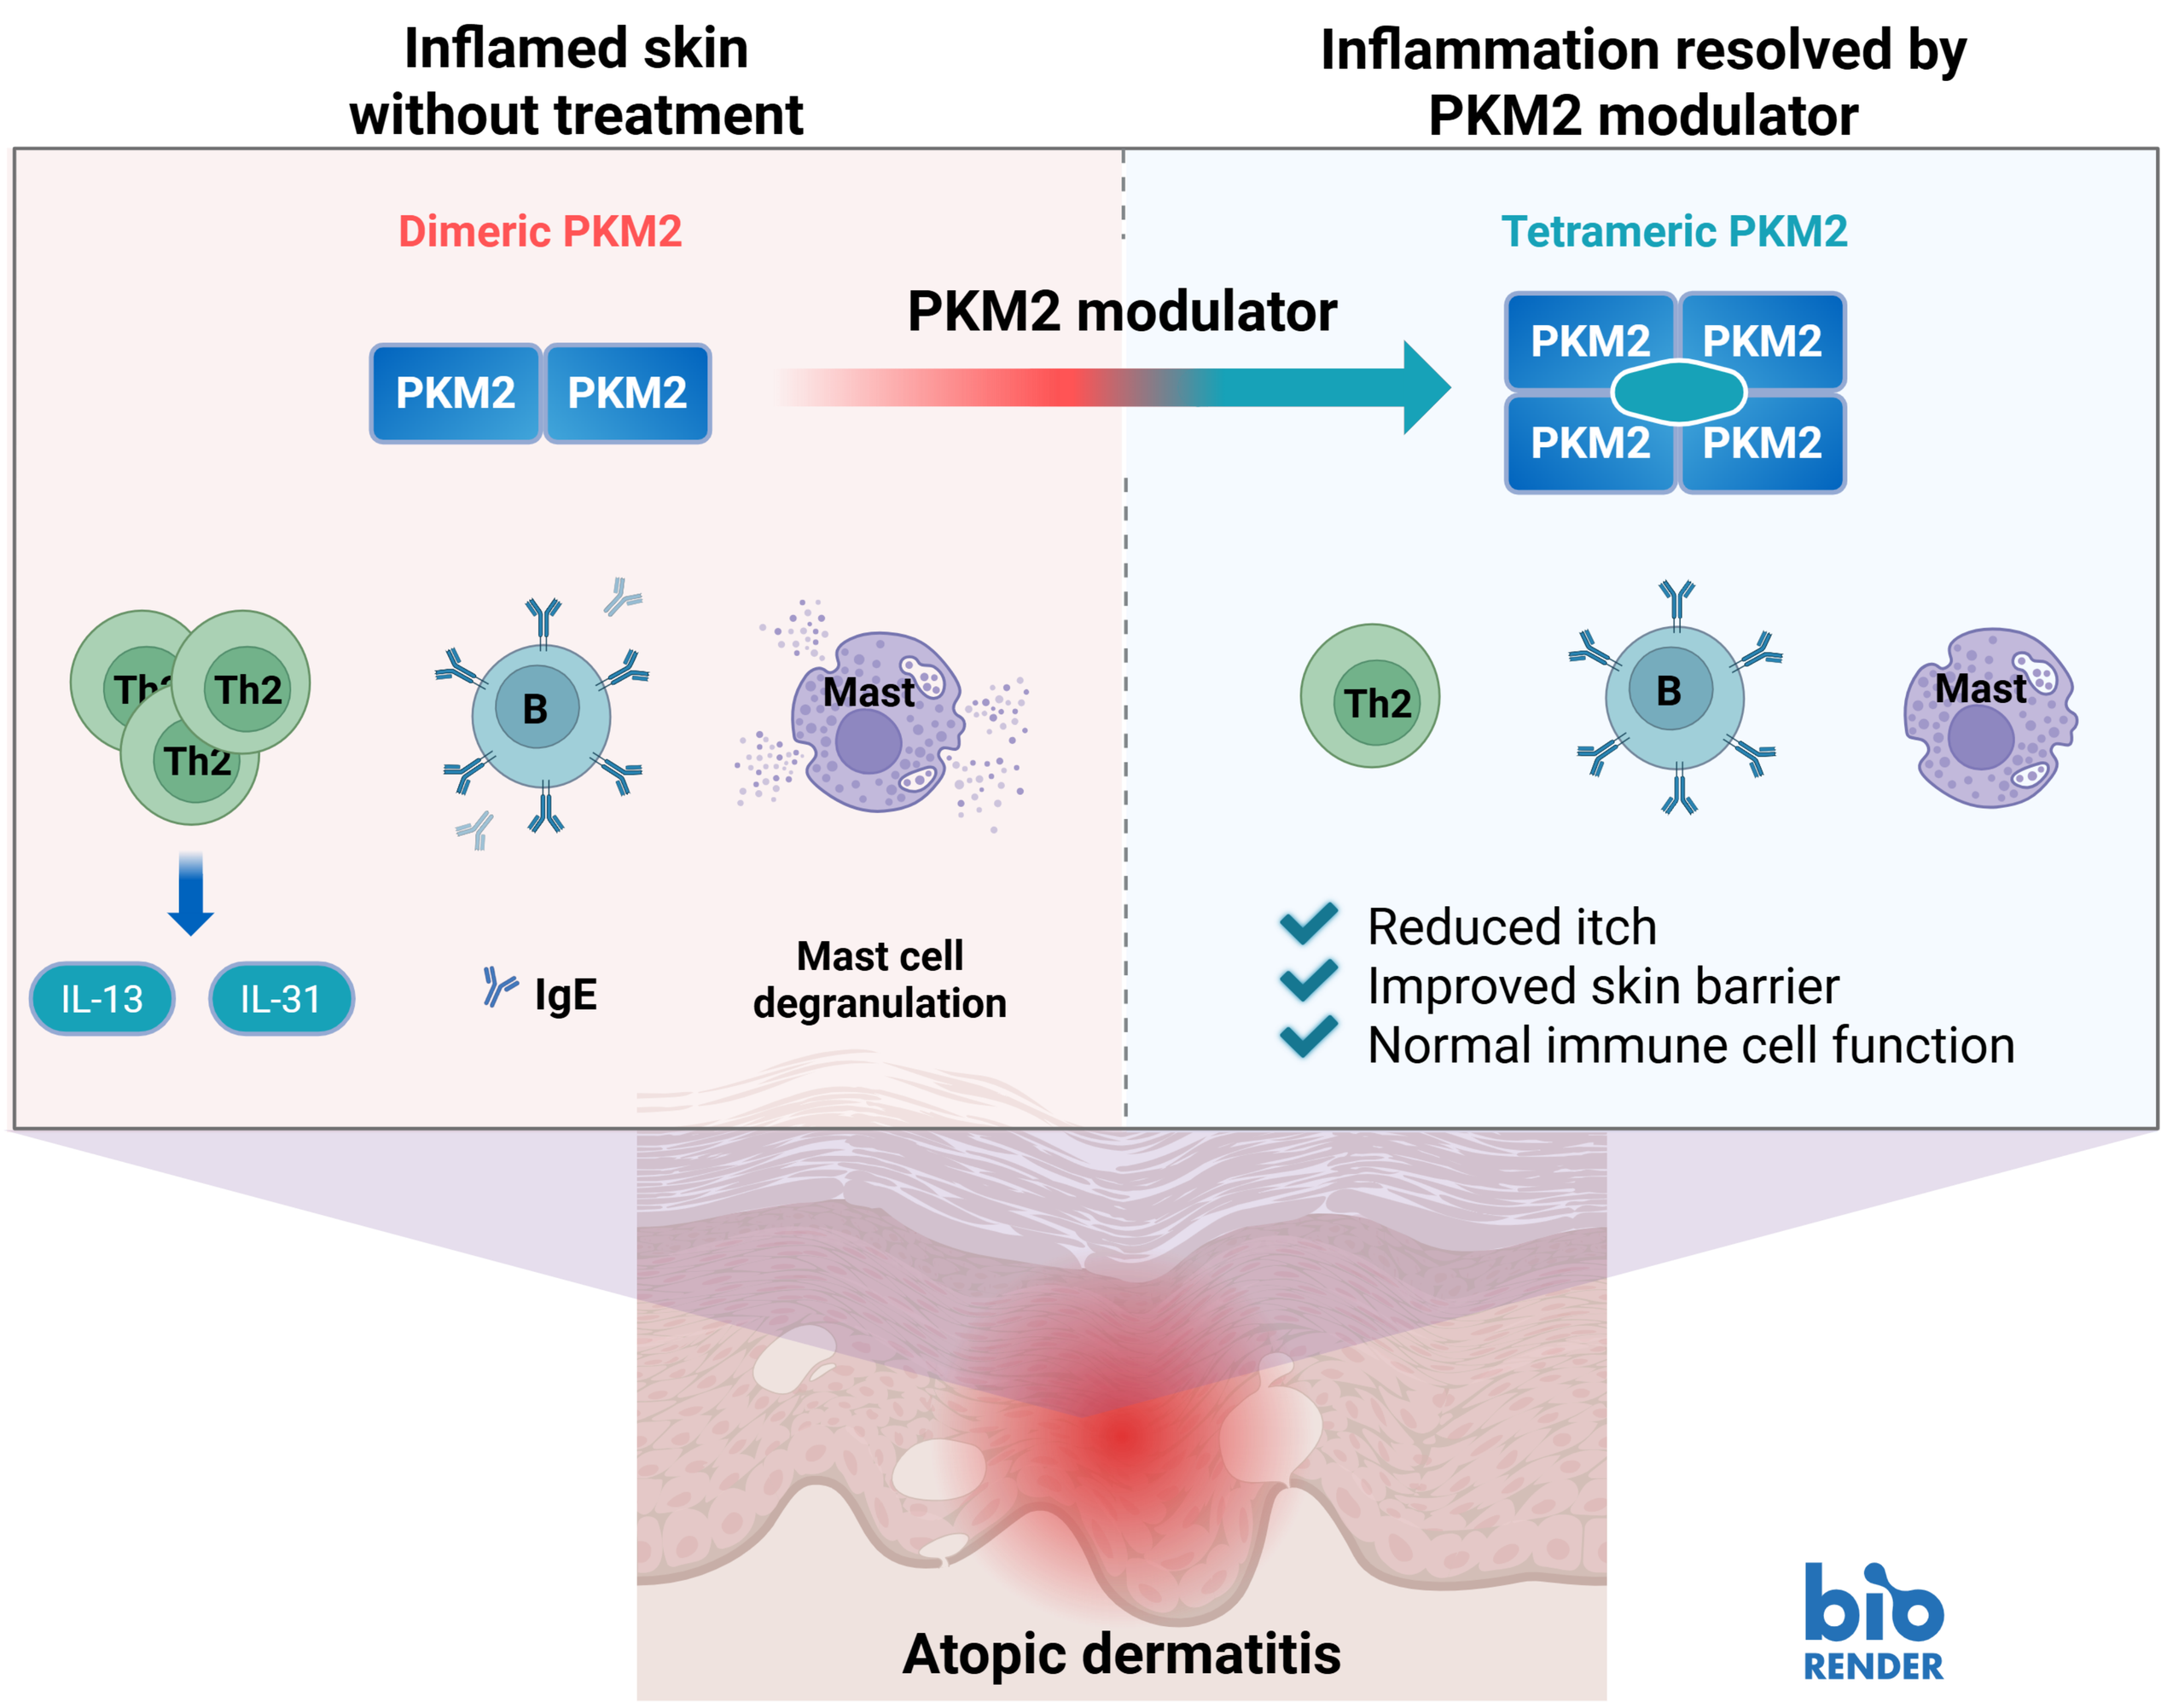
Inflamed skin without treatment
Inflammation resolved by PKM2 modulator
Dimeric PKM2
PKM2 PKM2
PKM2 modulator
Tetrameric PKM2
PKM2 PKM2 PKM2 PKM2
Th2 Th2 Th2
B
Mast
IL-13 IL-31
IgE
Mast cell degranulation
Reduced itch
Improved skin barrier
Normal immune cell function
Atopic dermatitis
bio RENDER

2. Induction of PKM2 in AD lesional tissue

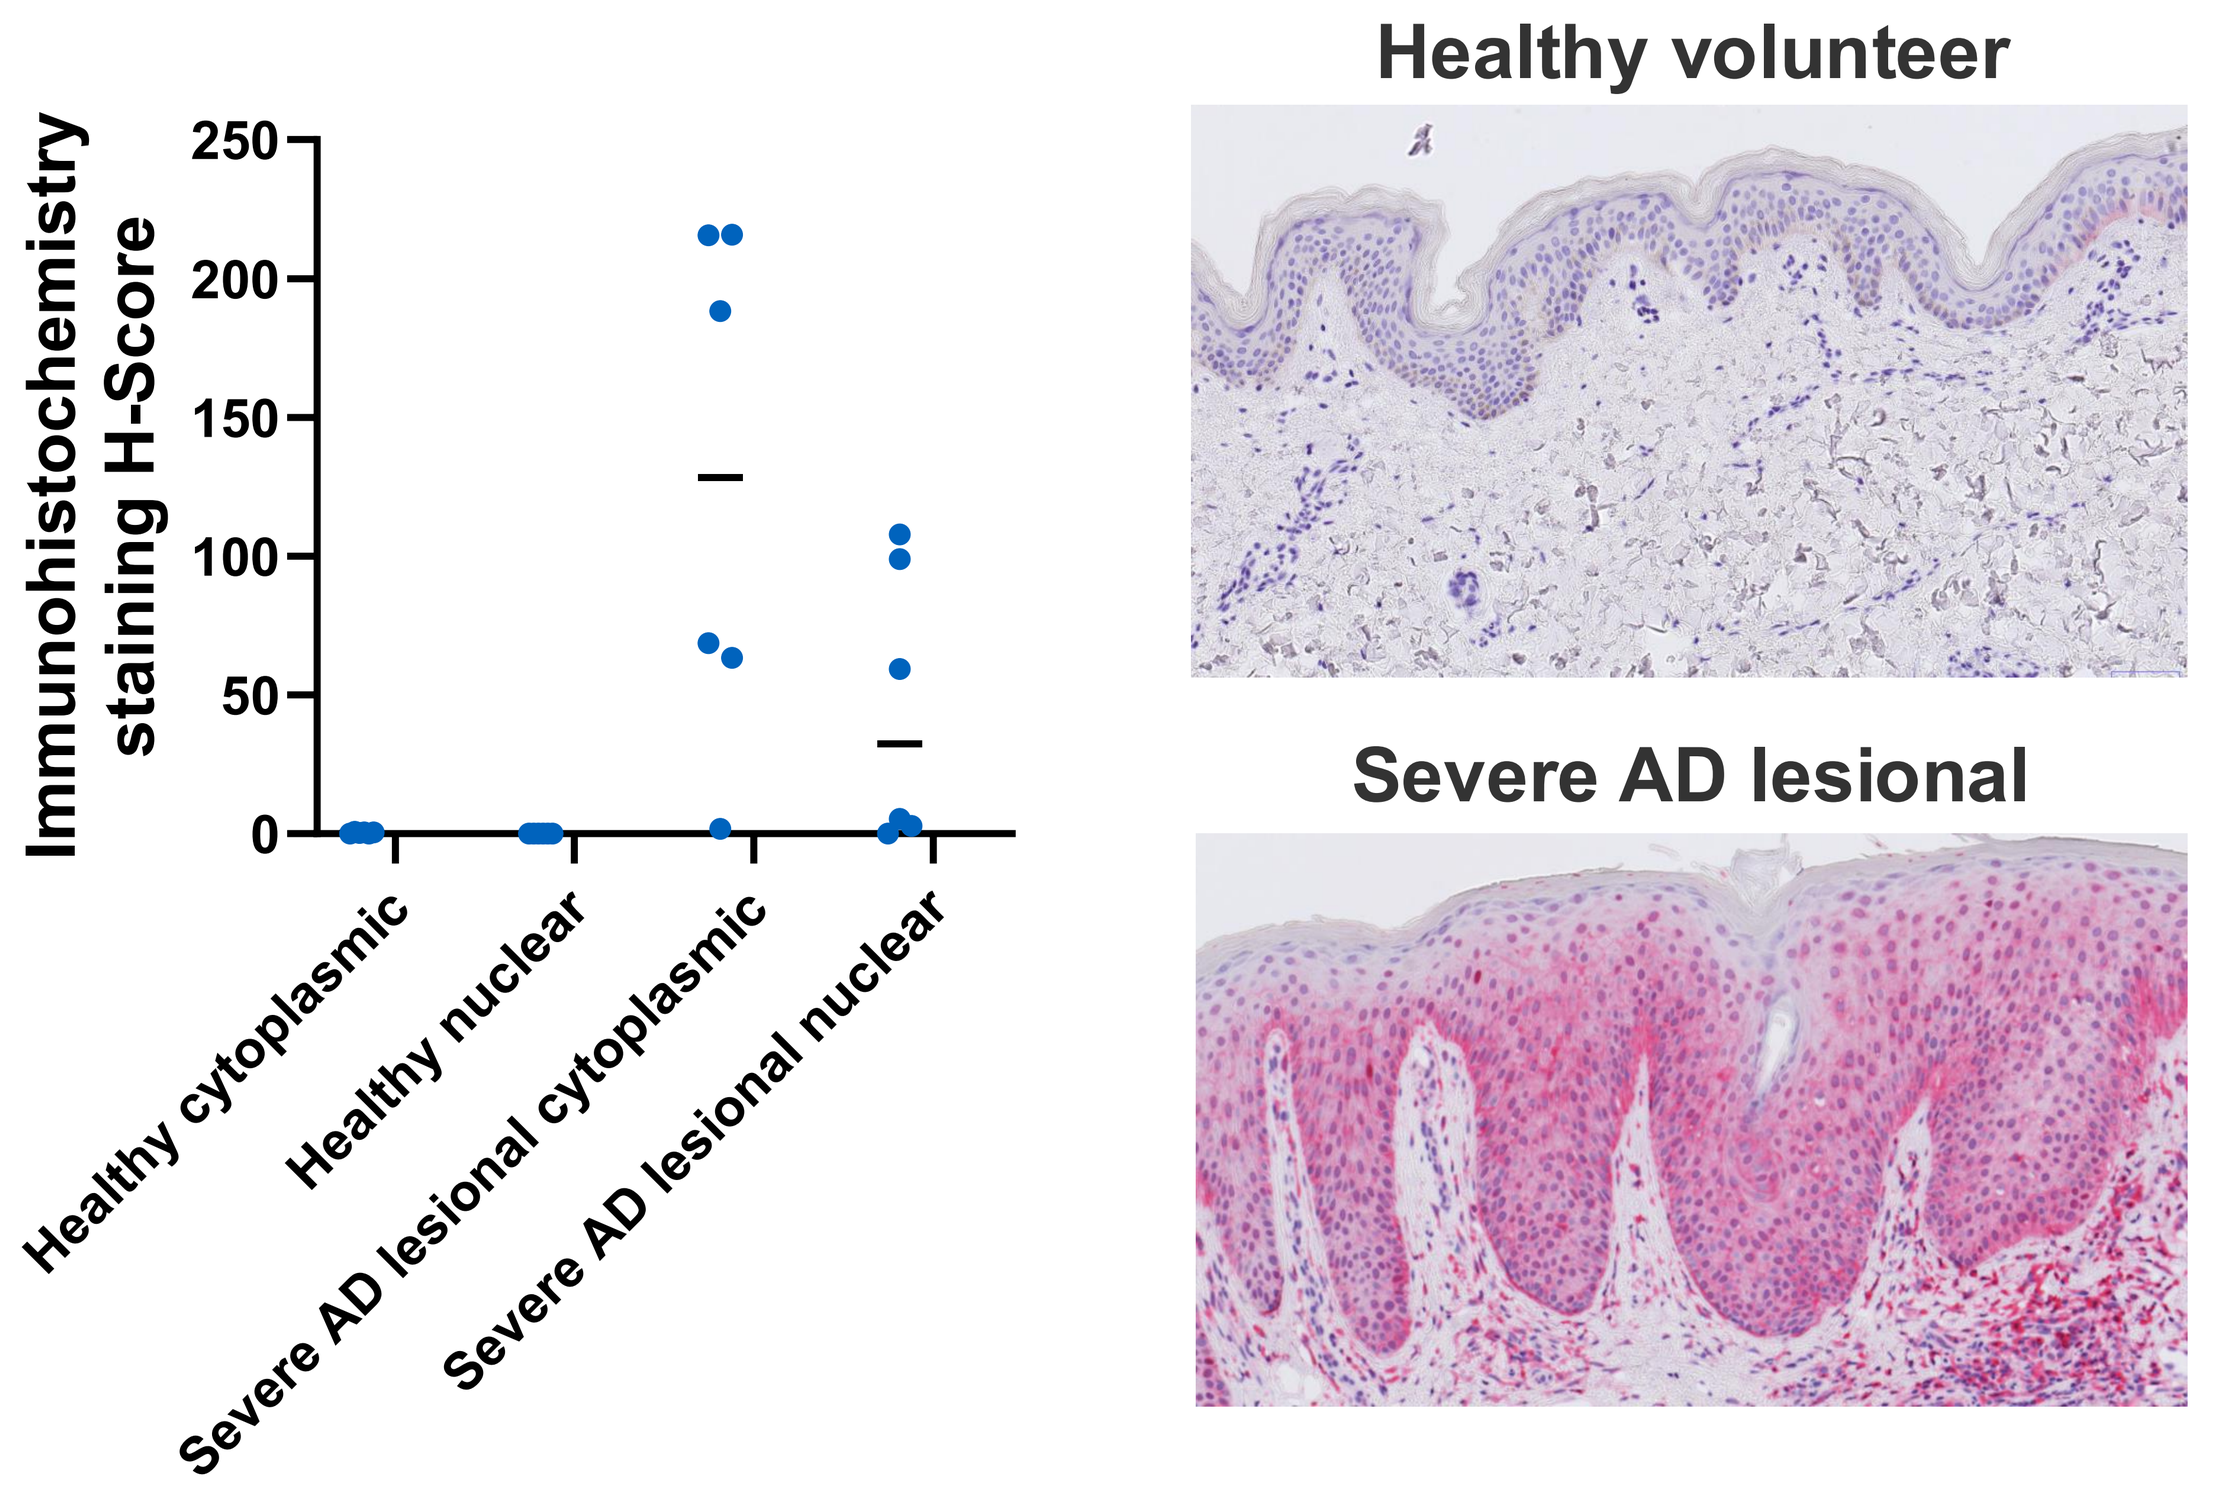
Immunohistochemistry staining H-Score
0 50 100 150 200 250
Healthy cytoplasmic
Healthy nuclear
Severe AD lesional cytoplasmic
Severe AD lesional nuclear
Healthy volunteer
Severe AD lesional

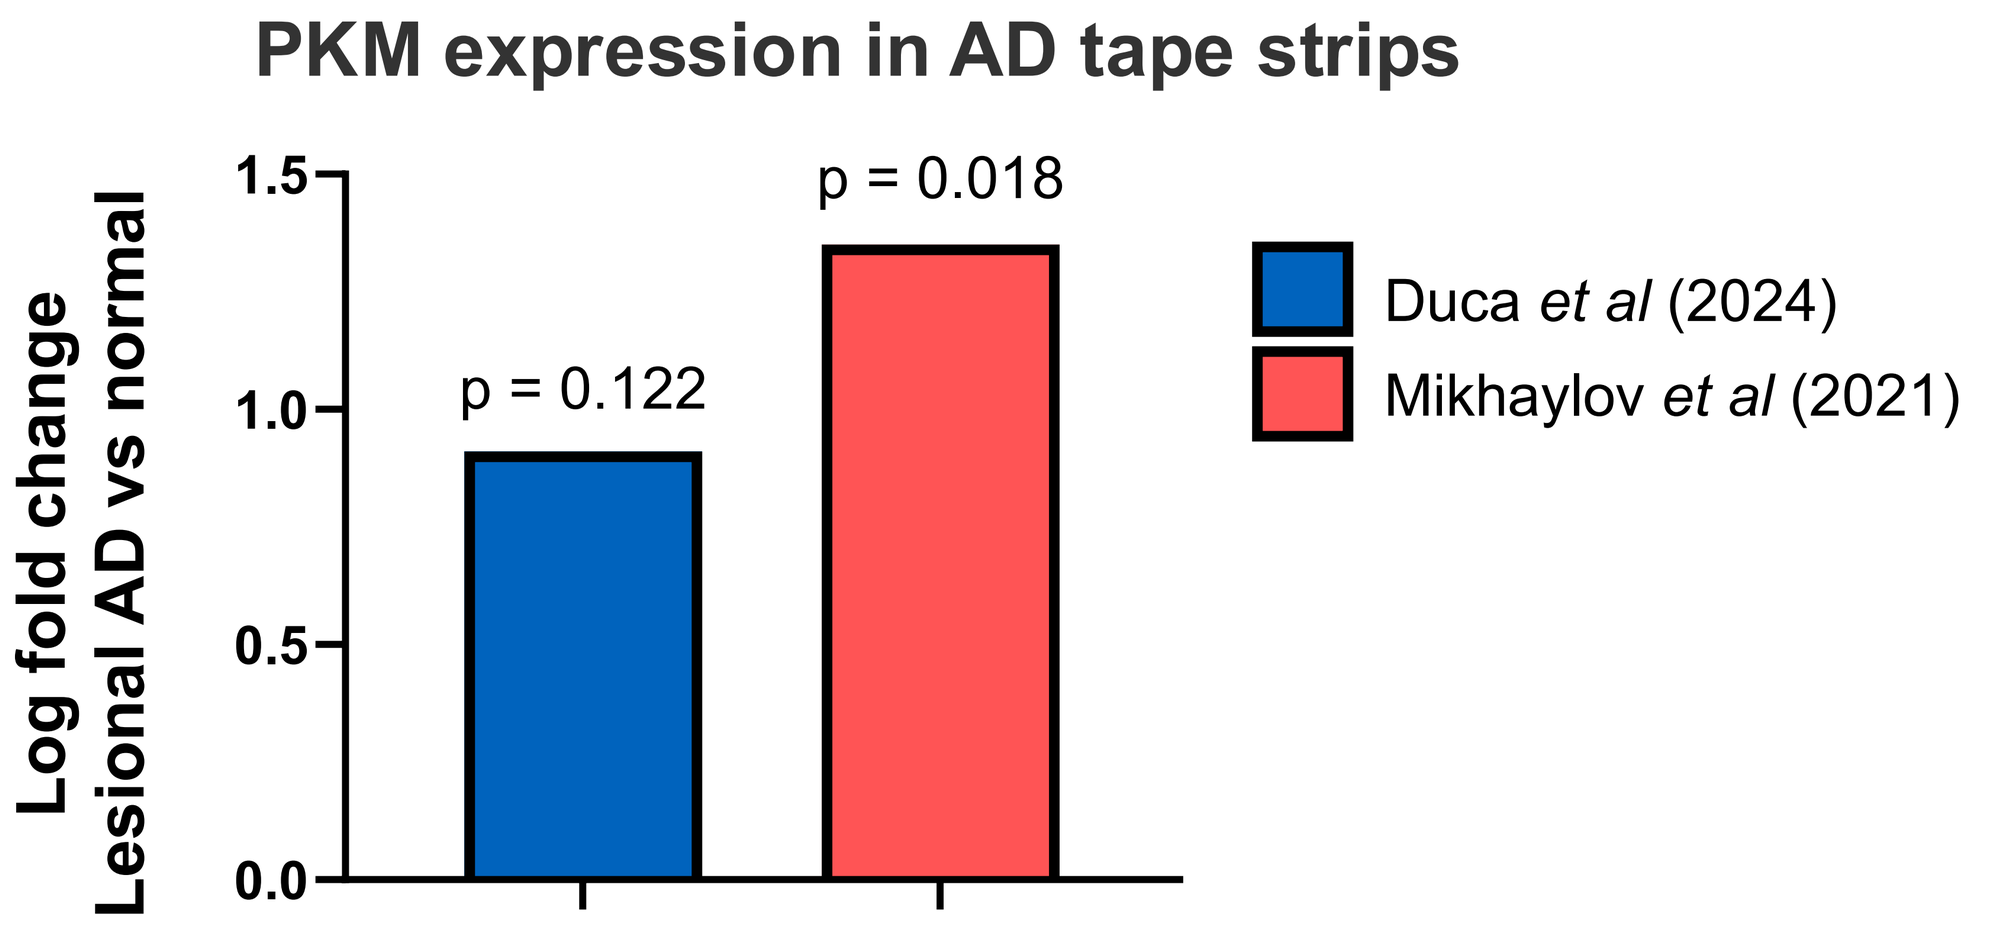
PKM expression in AD tape strips
Log fold change Lesional AD vs normal
0.0 0.5 1.0 1.5
p = 0.122
p = 0.018
Duca et al (2024)
Mikhaylov et al (2021)

3. PKM2 modulators activate PKM2

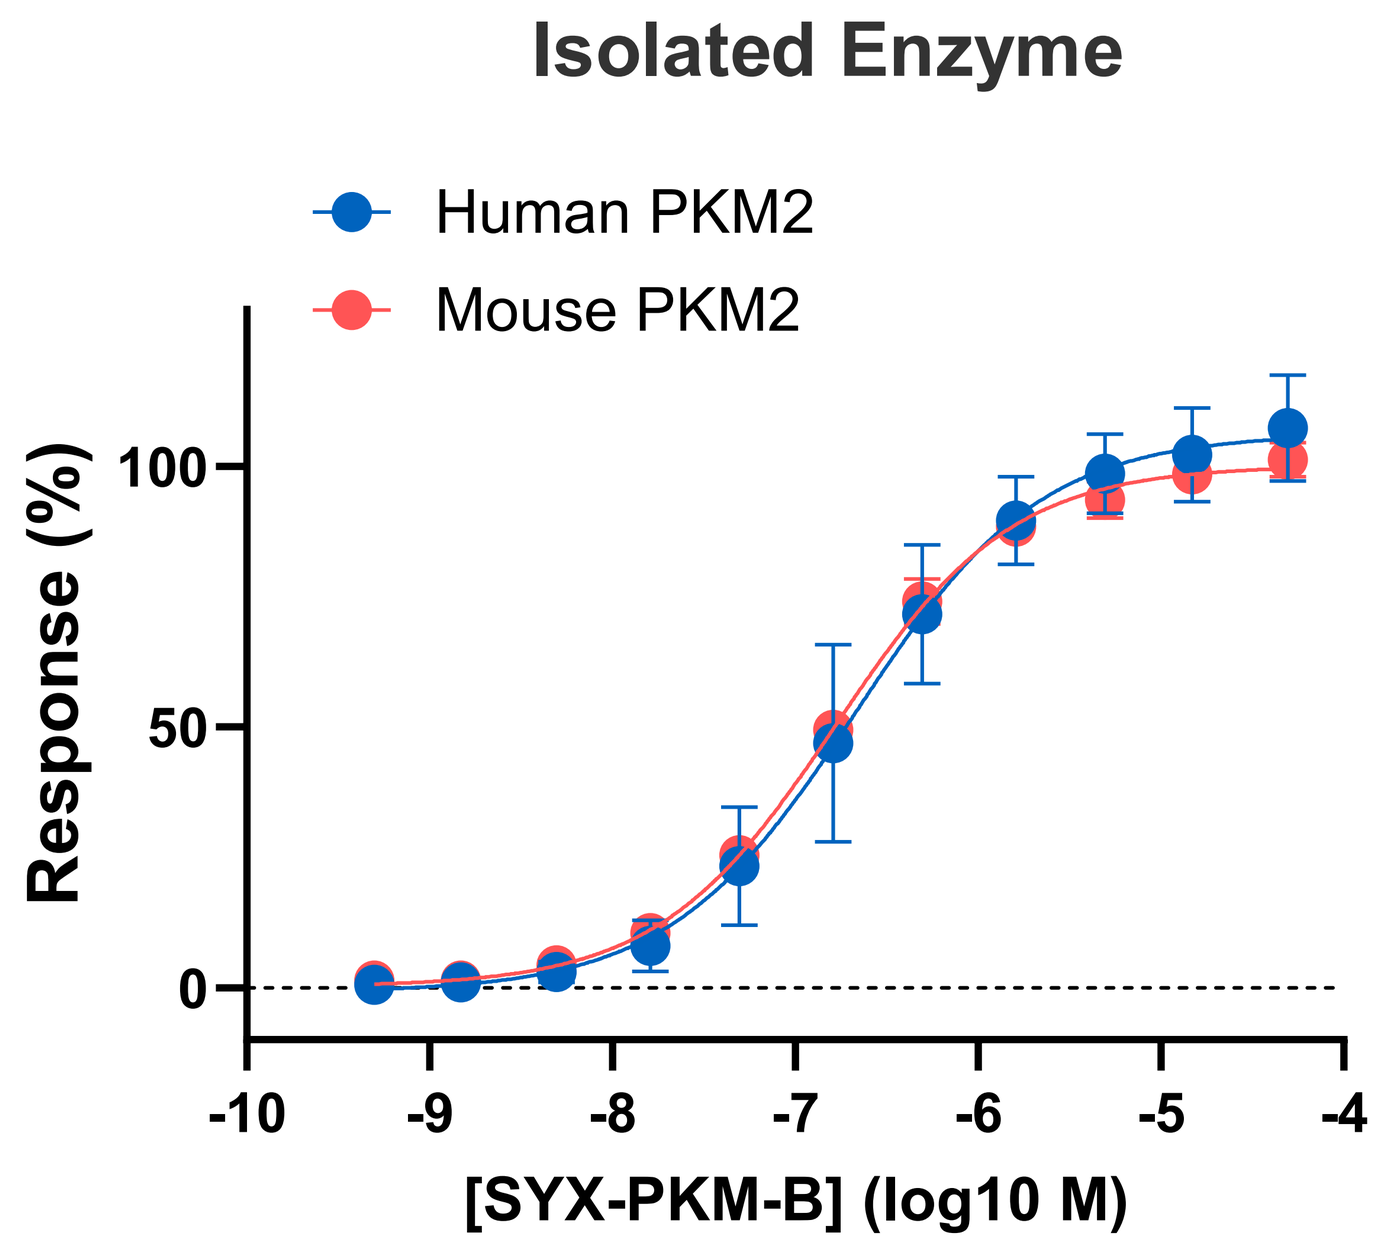
Isolated Enzyme
Human PKM2
Mouse PKM2
Response (%)
0 50 100
-10 -9 -8 -7 -6 -5 -4
[SYX-PKM-B] (log10 M)

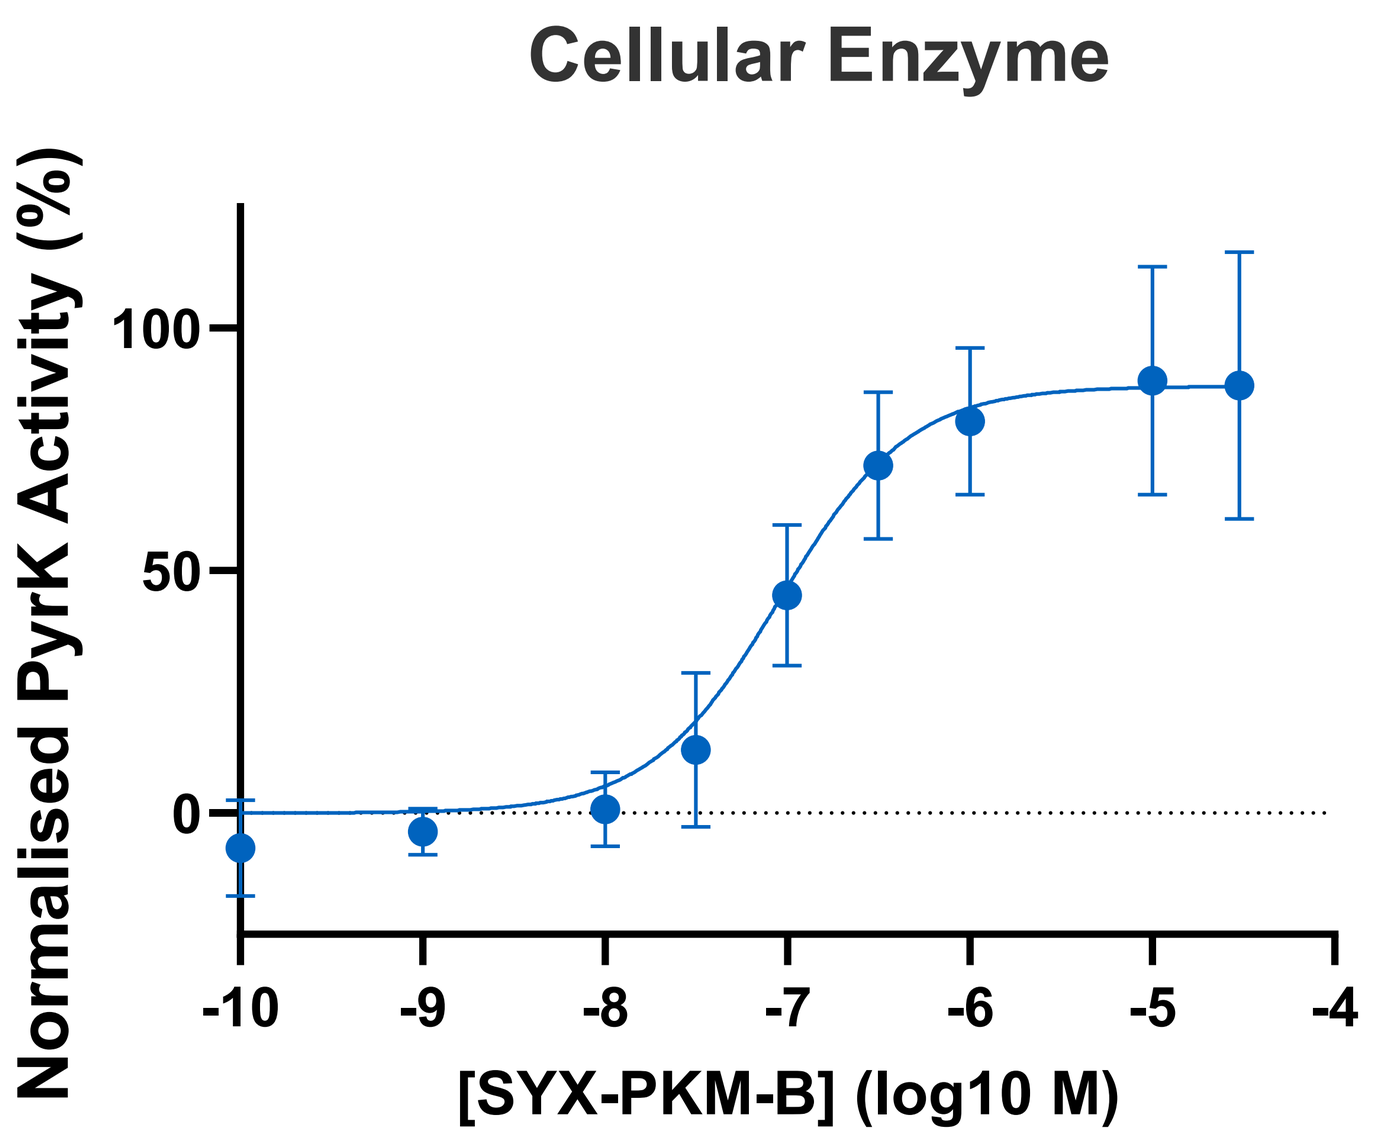
Cellular Enzyme
Normalised PyrK Activity (%)
0 50 100
-10 -9 -8 -7 -6 -5 -4
[SYX-PKM-B] (log10 M)

4. PKM2 modulators positively impact AD markers in a DNFB skin inflammation model

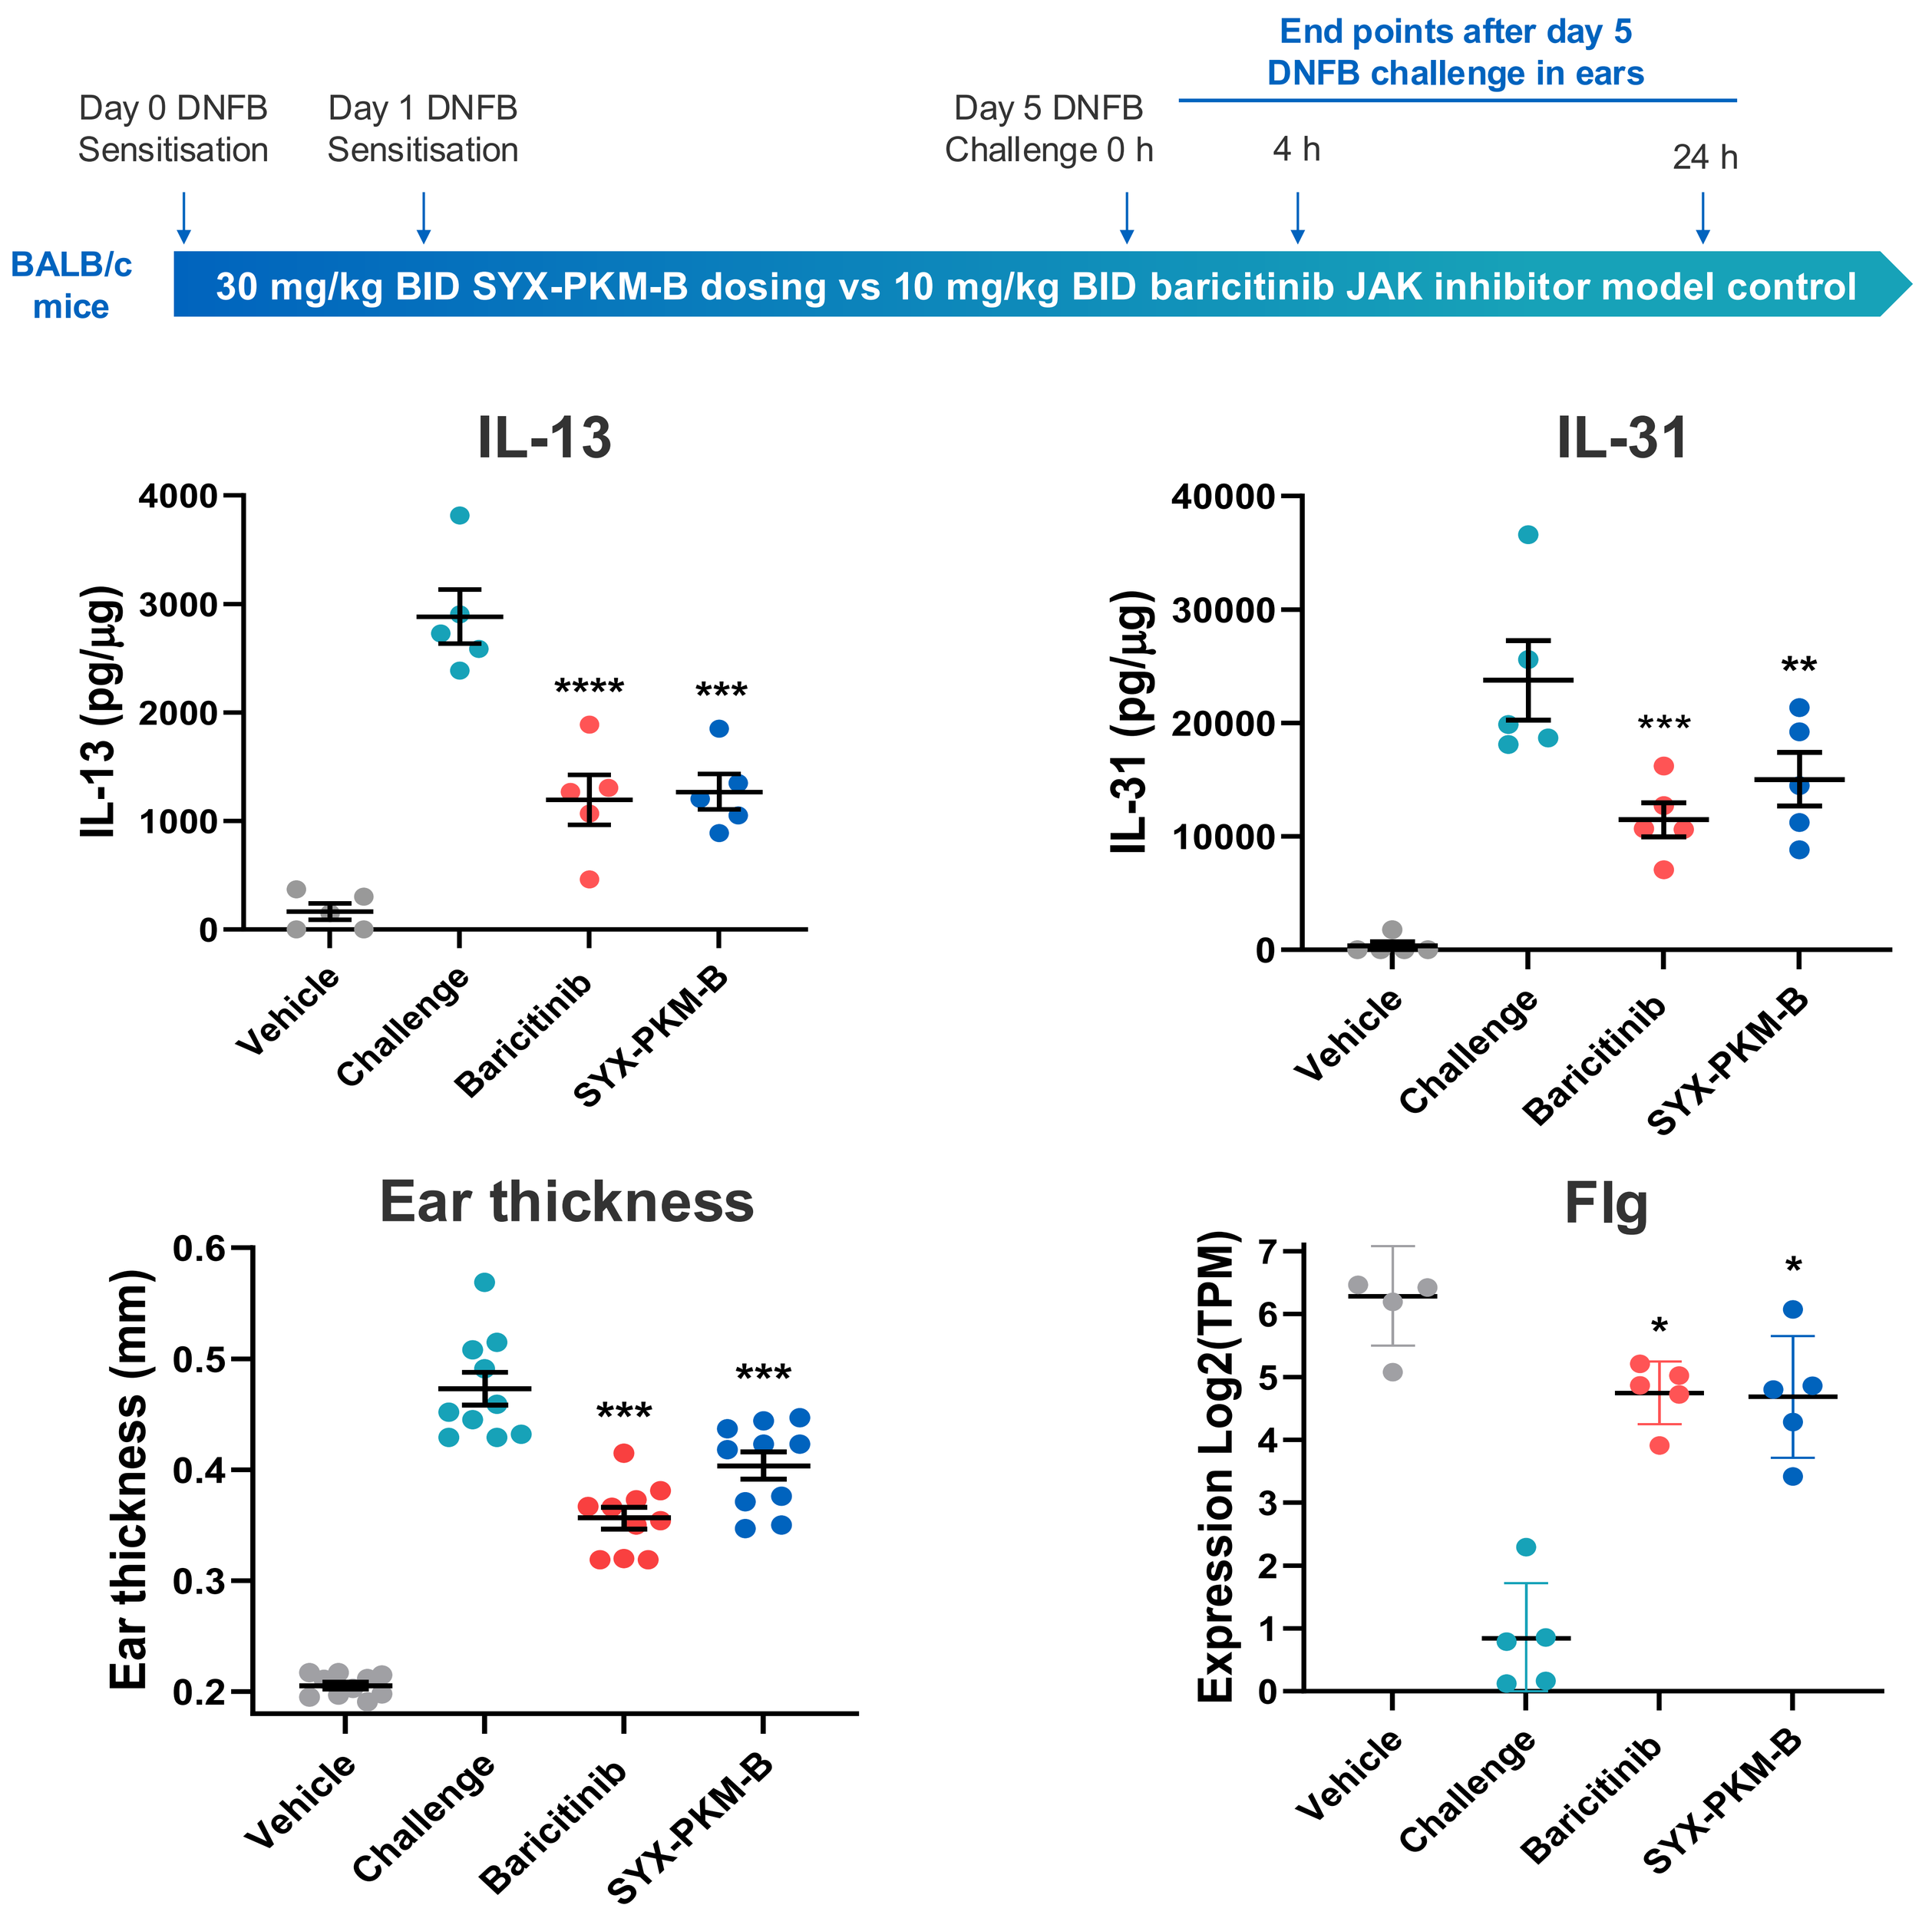
Day 0 DNFB Sensitisation
Day 1 DNFB Sensitisation
Day 5 DNFB Challenge 0 h
End points after day 5 DNFB challenge in ears
4 h
24 h
BALB/c mice
30 mg/kg BID SYX-PKM-B dosing vs 10 mg/kg BID baricitinib JAK inhibitor model control
IL-13
IL-13 (pg/µg)
IL-31
IL-31 (pg/µg)
Ear thickness
Ear thickness (mm)
Flg
Expression Log2(TPM)
Vehicle
Challenge
Baricitinib
SYX-PKM-B

5. PKM2 modulation positively impacts skin thickness and excoriations

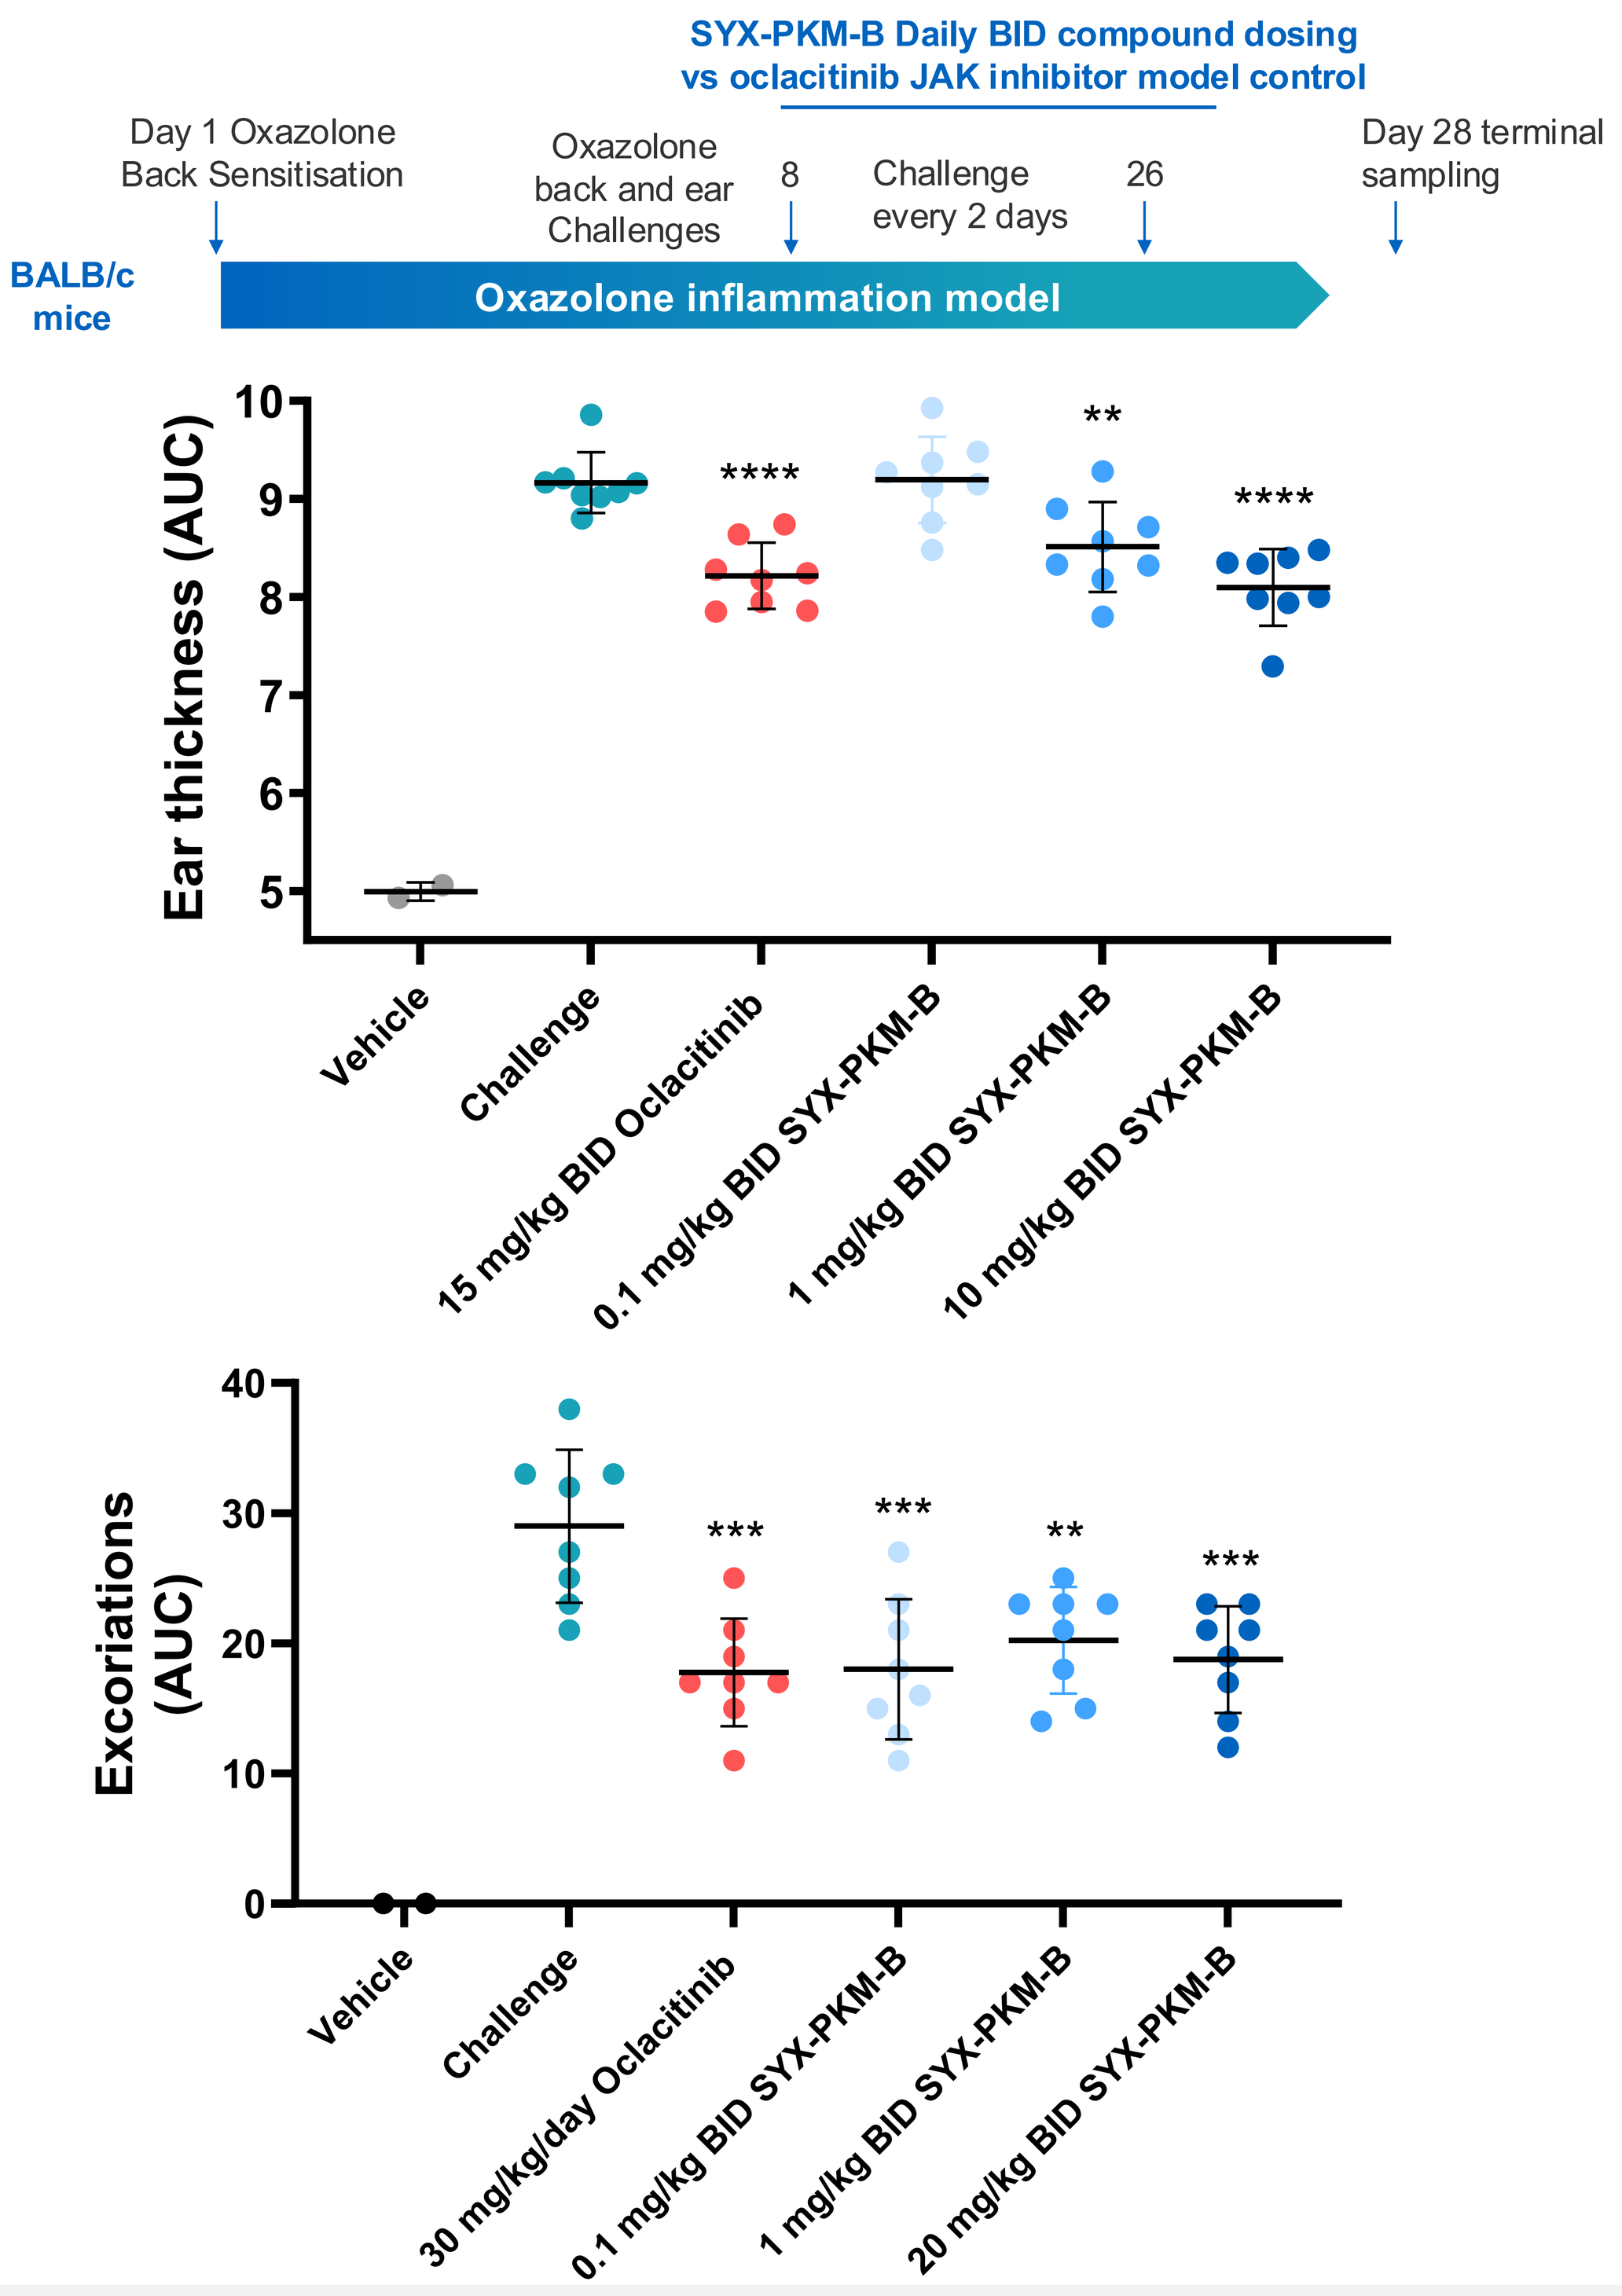
SYX-PKM-B Daily BID compound dosing vs oclacitinib JAK inhibitor model control
Day 1 Oxazolone Back Sensitisation
Oxazolone back and ear Challenges
8
Challenge every 2 days
26
Day 28 terminal sampling
BALB/c mice
Oxazolone inflammation model
Ear thickness (AUC)
Vehicle
Challenge
15 mg/kg BID Oclacitinib
0.1 mg/kg BID SYX-PKM-B
1 mg/kg BID SYX-PKM-B
10 mg/kg BID SYX-PKM-B
Excoriations (AUC)
30 mg/kg/day Oclacitinib
20 mg/kg BID SYX-PKM-B

6. PKM2 modulation inhibits mast cell degranulation

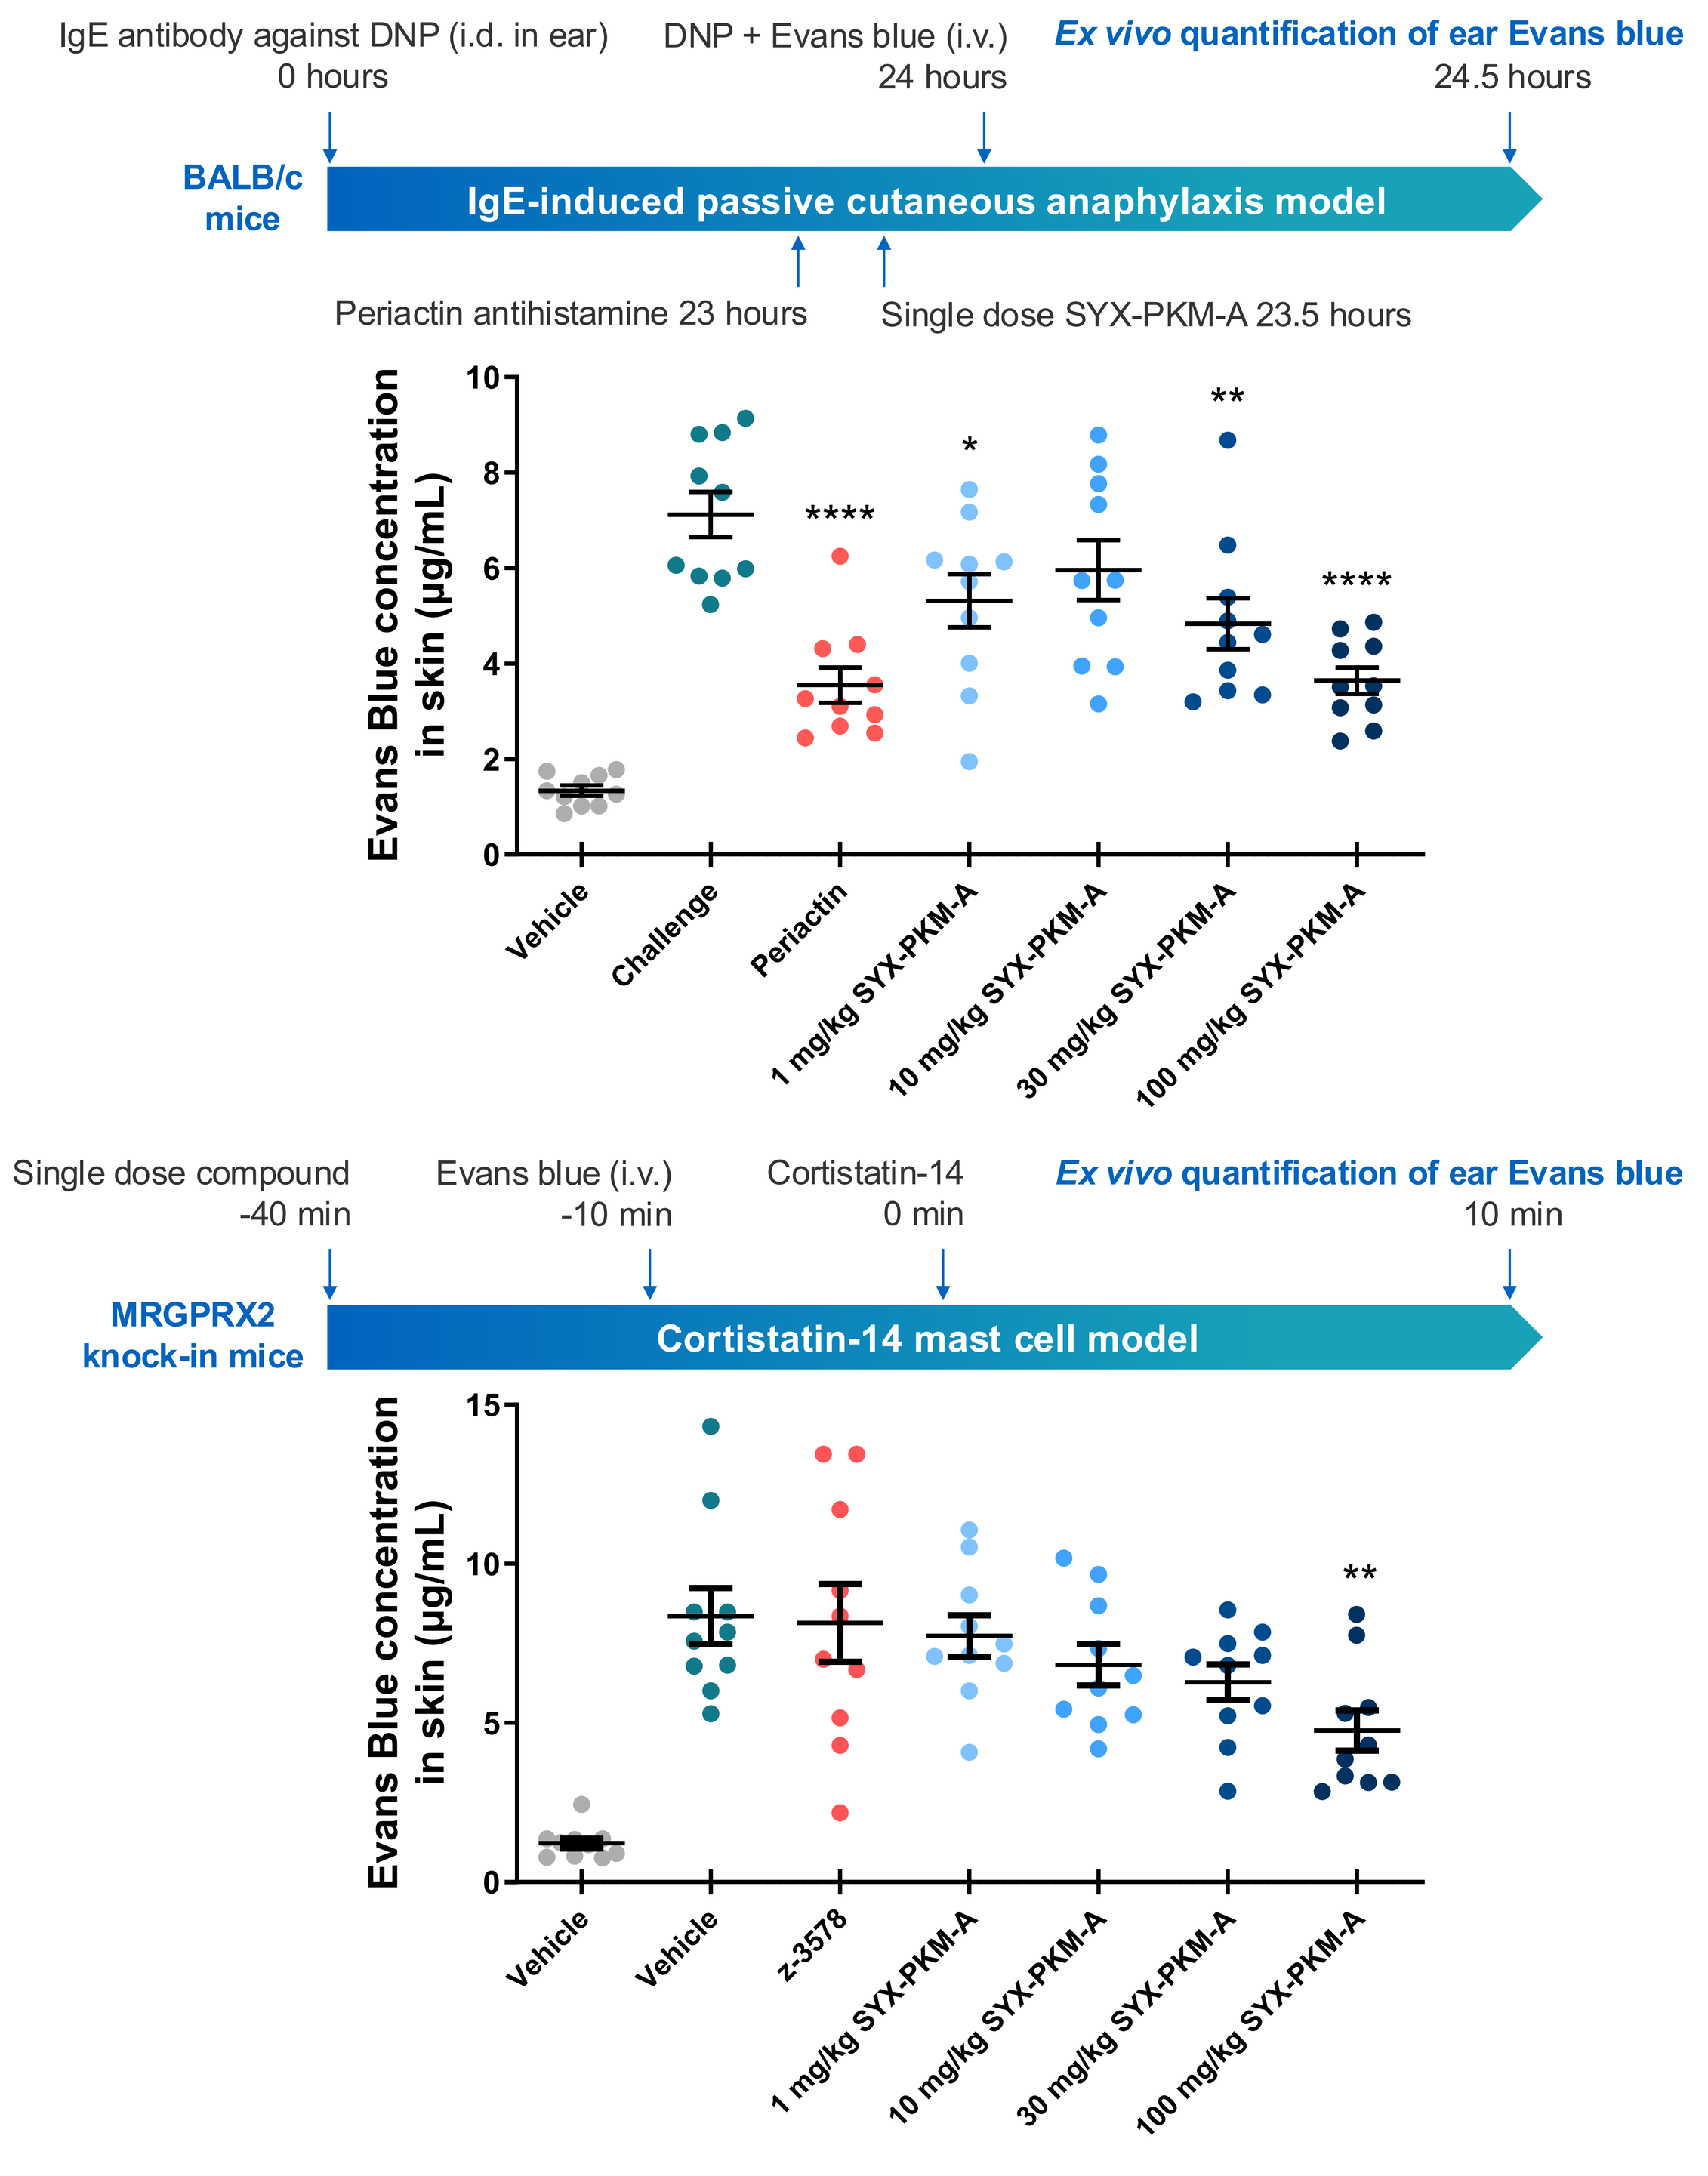
IgE antibody against DNP (i.d. in ear) 0 hours
DNP + Evans blue (i.v.) 24 hours
Ex vivo quantification of ear Evans blue 24.5 hours
BALB/c mice
IgE-induced passive cutaneous anaphylaxis model
Periactin antihistamine 23 hours
Single dose SYX-PKM-A 23.5 hours
Evans Blue concentration in skin (µg/mL)
Vehicle
Challenge
Periactin
1 mg/kg SYX-PKM-A
10 mg/kg SYX-PKM-A
30 mg/kg SYX-PKM-A
100 mg/kg SYX-PKM-A
Single dose compound -40 min
Evans blue (i.v.) -10 min
Cortistatin-14 0 min
Ex vivo quantification of ear Evans blue 10 min
MRGPRX2 knock-in mice
Cortistatin-14 mast cell model
z-3578

1. E. D. Duca *et al.*, *Allergy*. **79**, 80–92 (2024).
2. D. Mikhaylov *et al.*, *Dermatitis*. **32**, S71–S80 (2021).

Conclusions

PKM2 is overexpressed in lesional AD skin. Novel small molecule PKM2 modulators, developed as a potential AD therapeutic, activate PKM2 in isolated enzyme and cellular assays. PKM2 modulation impacts inflammation, itch and barrier function in several murine models of inflammatory skin diseases. Furthermore, PKM2 modulation decreases mast cell degranulation in IgE-induced and cortistatin-14 induced mast cell models. Taken together, the studies show that PKM2 modulation inhibits several important mechanisms underlying AD pathology. PKM2 modulator, SYX-5219, has recently entered clinical trials in AD (Scan QR code). PKM2 modulation offers an oral, targeted, disease-modifying anti-inflammatory approach, targeting a critical enzyme that regulates cell metabolism. This PKM2 modulator could address the major unmet needs of AD where there are limited treatment options under the current standard of care.